CRISPR/Cas9 editing by NTLA-2002 remains durable in huKLKB1 mice following partial hepatectomy

[1]**Intellia Therapeutics,** Cambridge, MA, [2]**Regeneron Pharmaceuticals,** Tarrytown, NY

Authors
K. Walsh[1], P. Cottingham[1], J. Seitzer[1], C. Shaw[1], C. Blanchard[1], N. Gardner[1], V. Doshi[1], K. Wood[1], S. Soukamneuth[1], J. Phillips[1], B. Han[1], S. Jaligama[1], R. Cole[1], S. Parthasarathy[1], D. Kryukov[1], J. Butler[1], D. Kajimura[2], CJ Siao[2]

#507

Intellia Therapeutics' Platform for *In Vivo* Hepatic Gene Editing

In Vivo
CRISPR is the therapy

GENETIC DISEASES

- Potential curative therapy from single dose
- Systemic non-viral delivery of CRISPR/Cas9 provides transient expression and potential safety advantages
- Permanent gain of function with targeted gene insertion
- Capable of delivering to multiple tissue types for various therapeutic applications

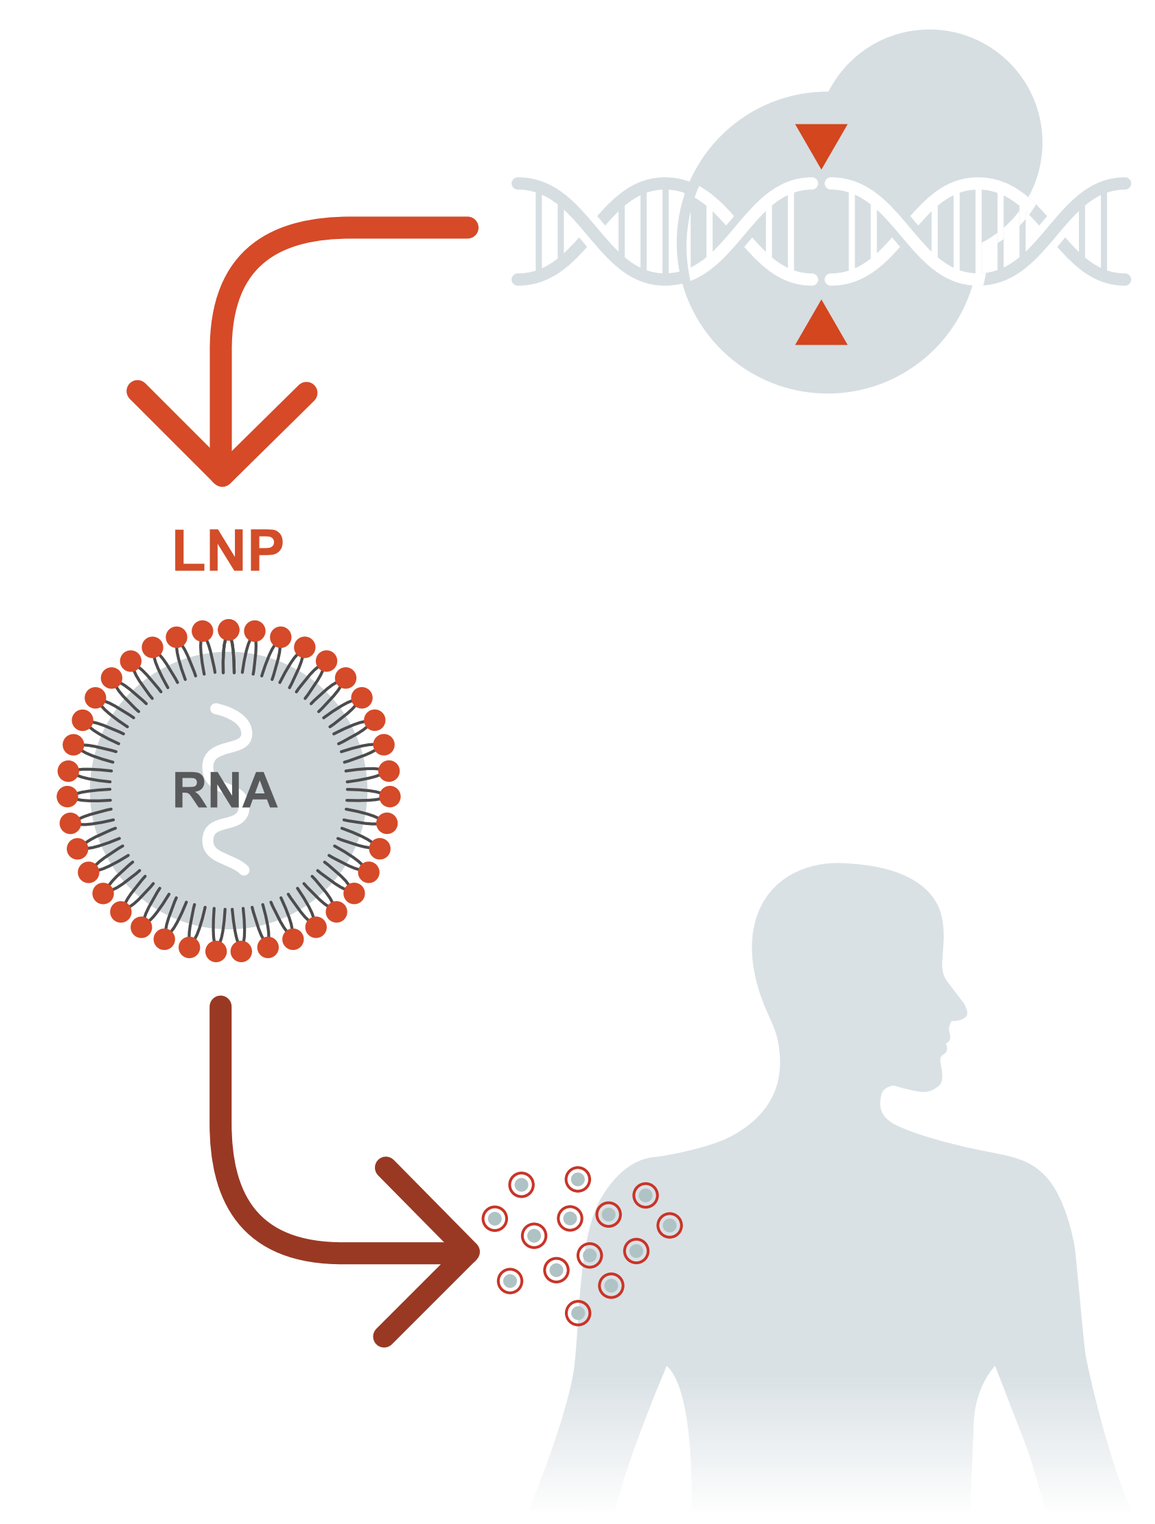
LNP
RNA

Modular Delivery Platform Enables Rapid and Reproducible Path to Clinical Development

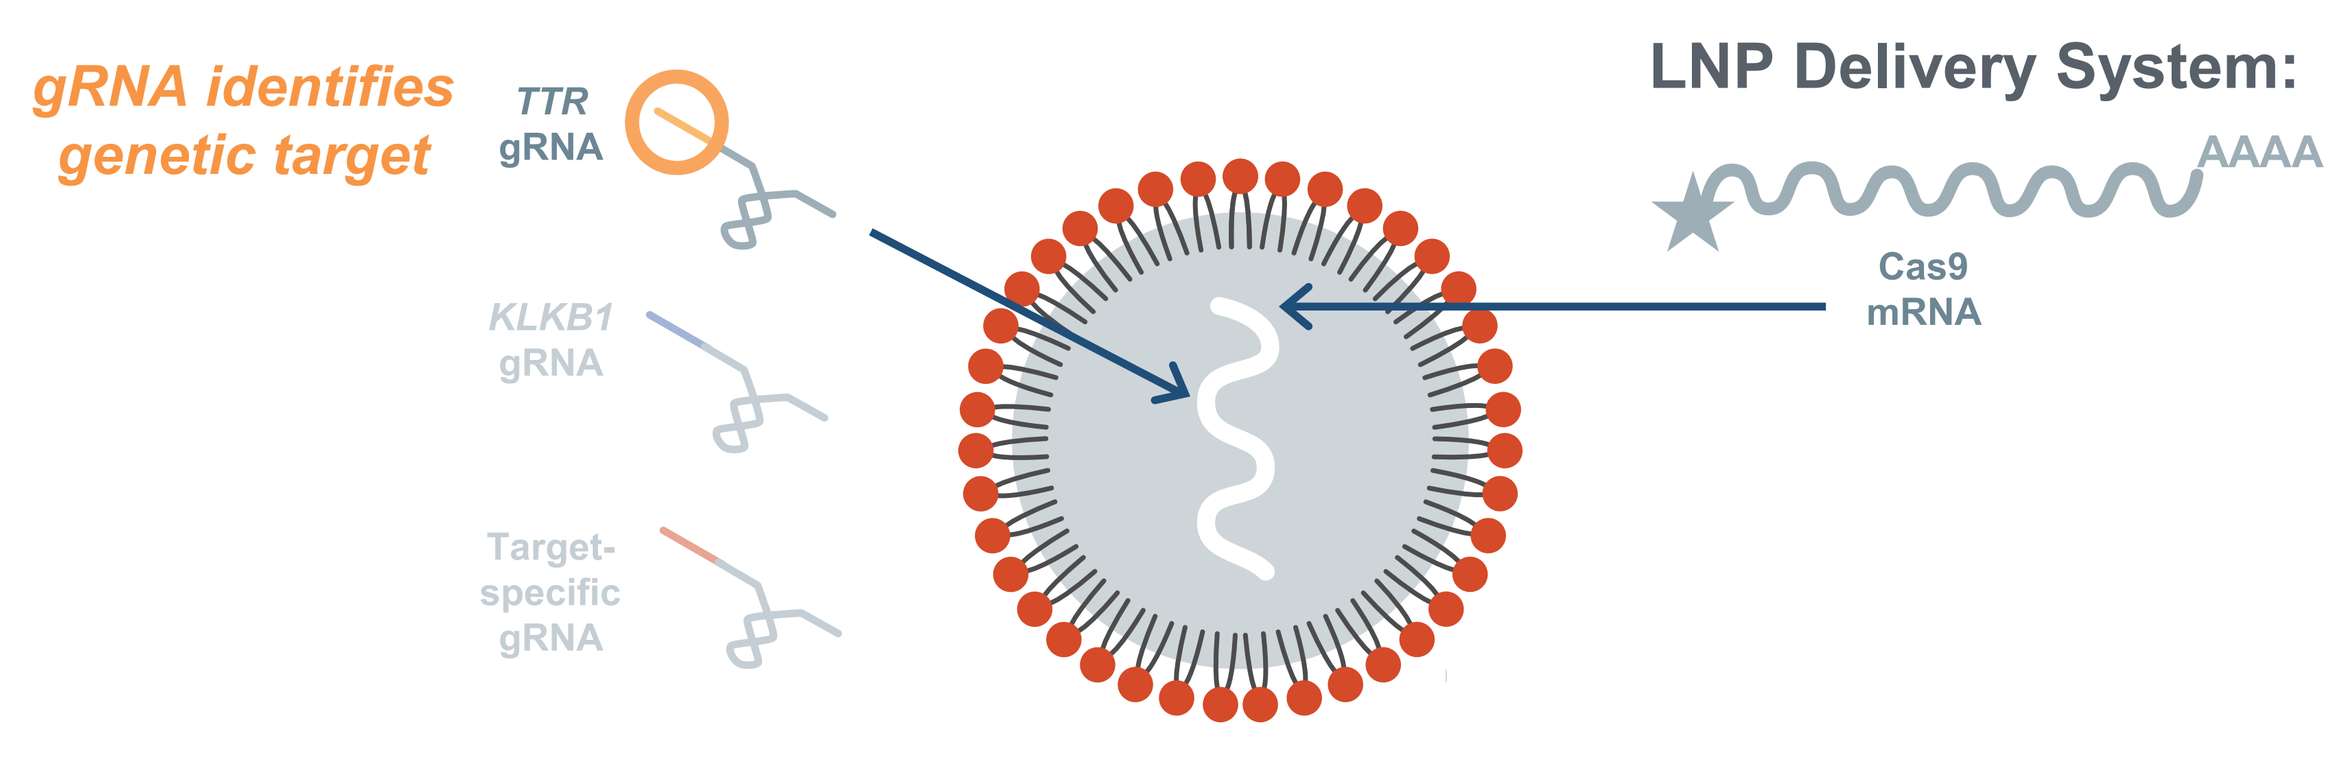
gRNA identifies genetic target
TTR gRNA
KLKB1 gRNA
Target-specific gRNA
LNP Delivery System:
AAAA
Cas9 mRNA

Key Advantages of LNP Delivery

- Clinically-proven delivery to liver
- Large cargo capacity
- Transient expression
- Biodegradable
- Low immunogenicity
- Well-tolerated
- Redosing capability
- Scalable synthetic manufacturing
- Tunable to other tissues

NTLA-2002 for HAE Prophylaxis

HAE Pathophysiology: Dysregulated Bradykinin Signaling Cascade Mediates HAE Attacks

C1-INH deficiency accounts for vast majority of HAE prevalence

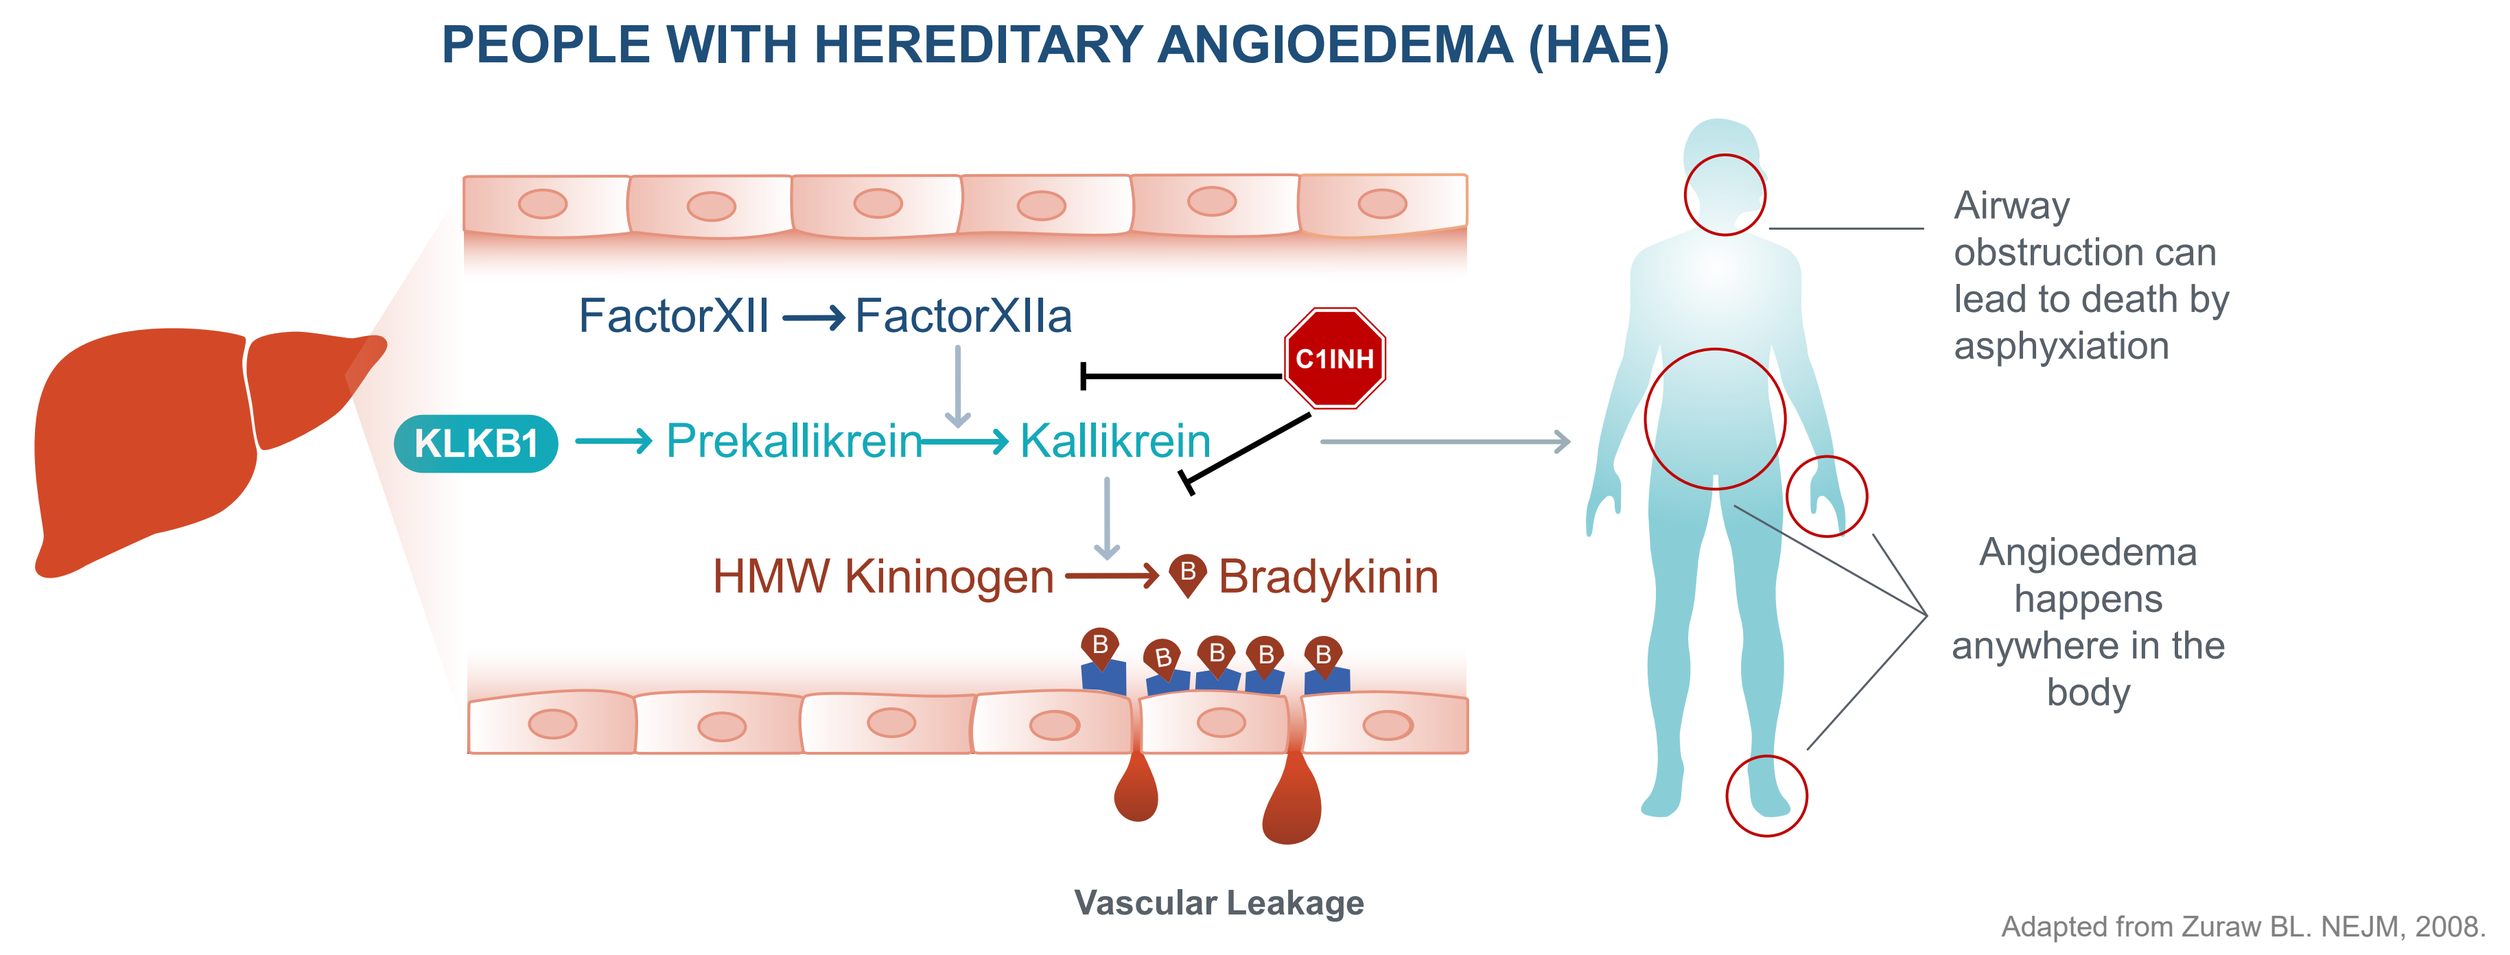
PEOPLE WITH HEREDITARY ANGIOEDEMA (HAE)
FactorXII → FactorXIIa
C1INH
KLKB1 → Prekallikrein → Kallikrein
HMW Kininogen → Bradykinin
Vascular Leakage
Airway obstruction can lead to death by asphyxiation
Angioedema happens anywhere in the body

Adapted from Zuraw BL. NEJM, 2008.

NTLA-2002 Therapeutic Hypothesis

Stable HAE prophylaxis by permanent disruption of *KLKB1* using CRISPR/Cas9 gene editing

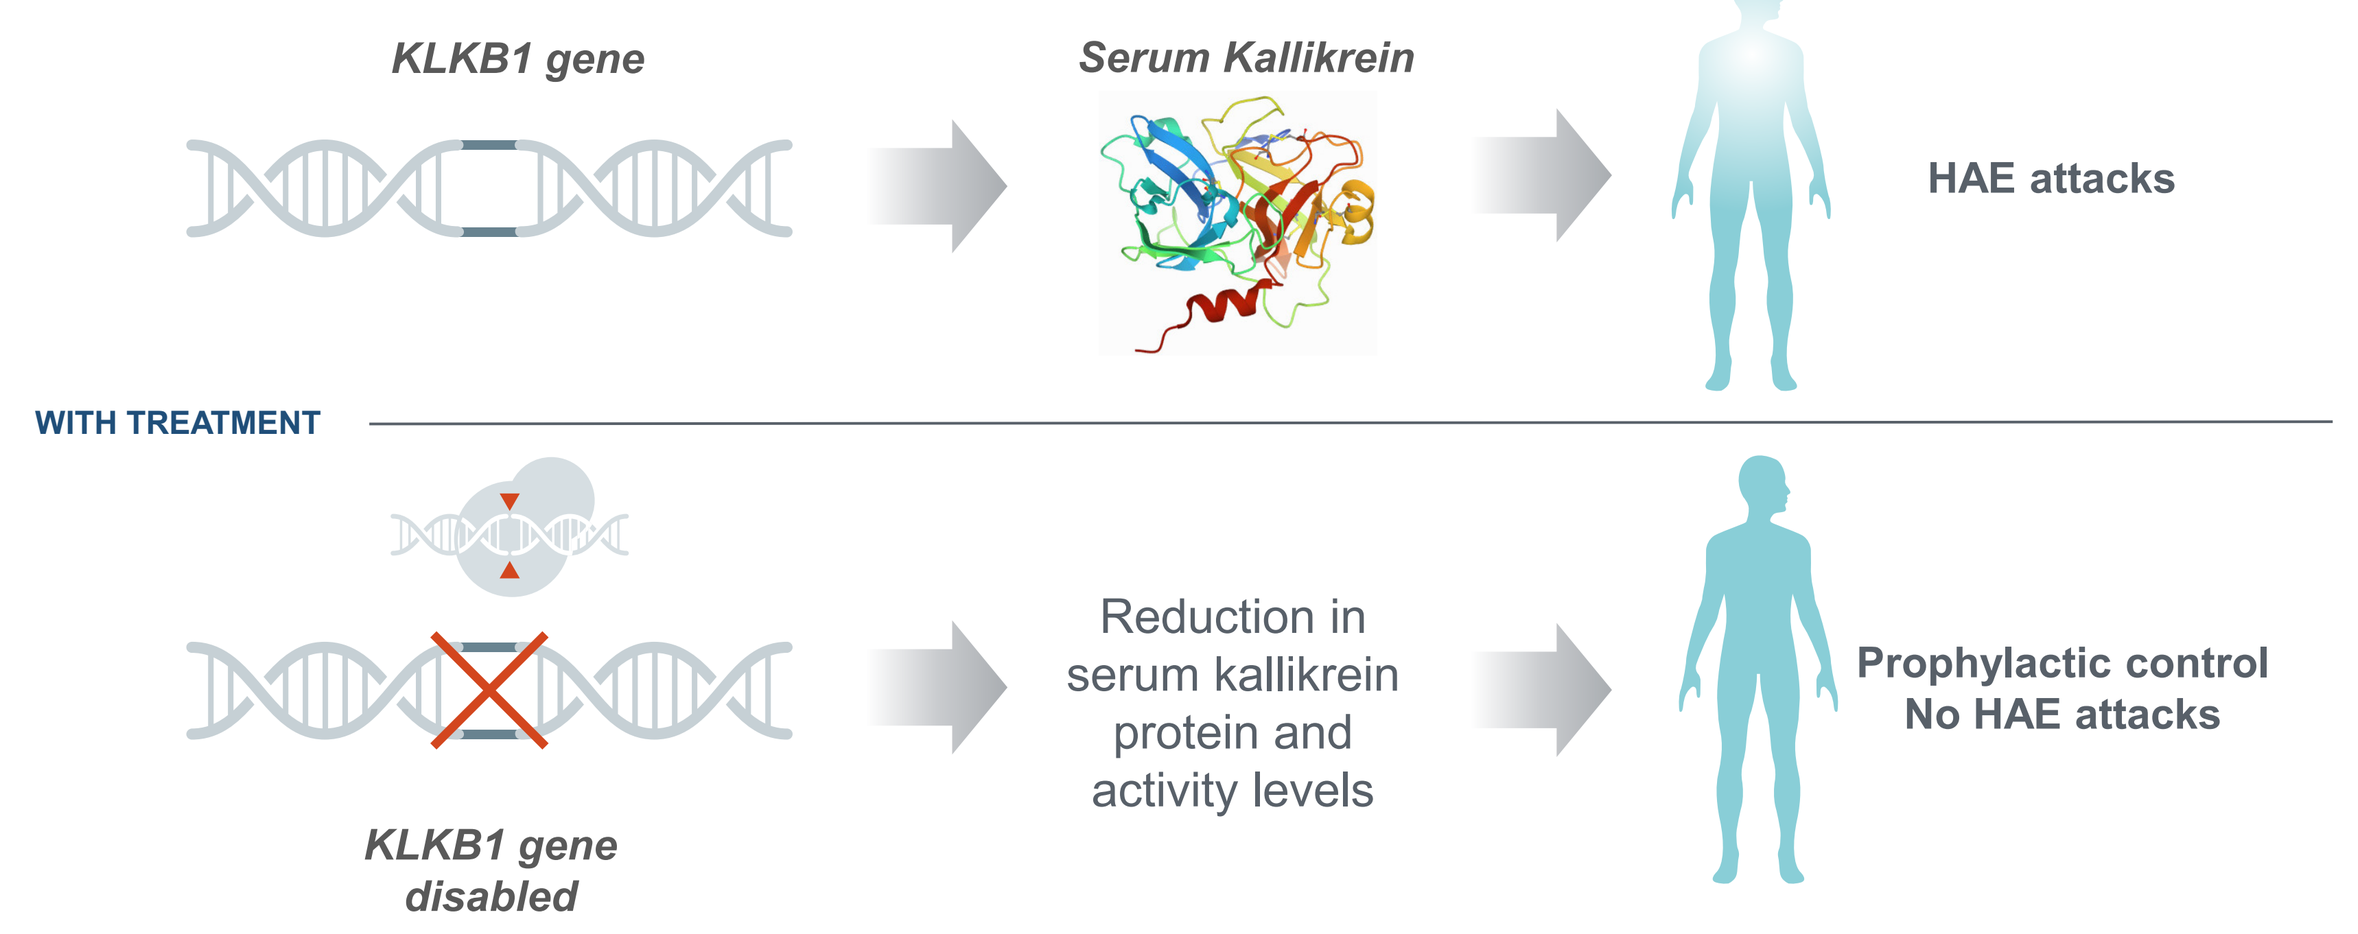
KLKB1 gene
Serum Kallikrein
HAE attacks
WITH TREATMENT
Reduction in serum kallikrein protein and activity levels
Prophylactic control
No HAE attacks
KLKB1 gene disabled

Adapted from Partridge et al., J. Struct. Bio, 2019

Rationale

- Long-term durability studies in mice treated with NTLA-2002 show sustained gene editing over 12-months demonstrating that CRISPR/Cas9-mediated gene edits persist through regular cell turnover.
- This study examines the durability of gene editing in hepatocytes with NTLA-2002 using a surgical rodent model, a partial hepatectomy (PHx), where 70% of the liver is removed and allowed to regenerate.

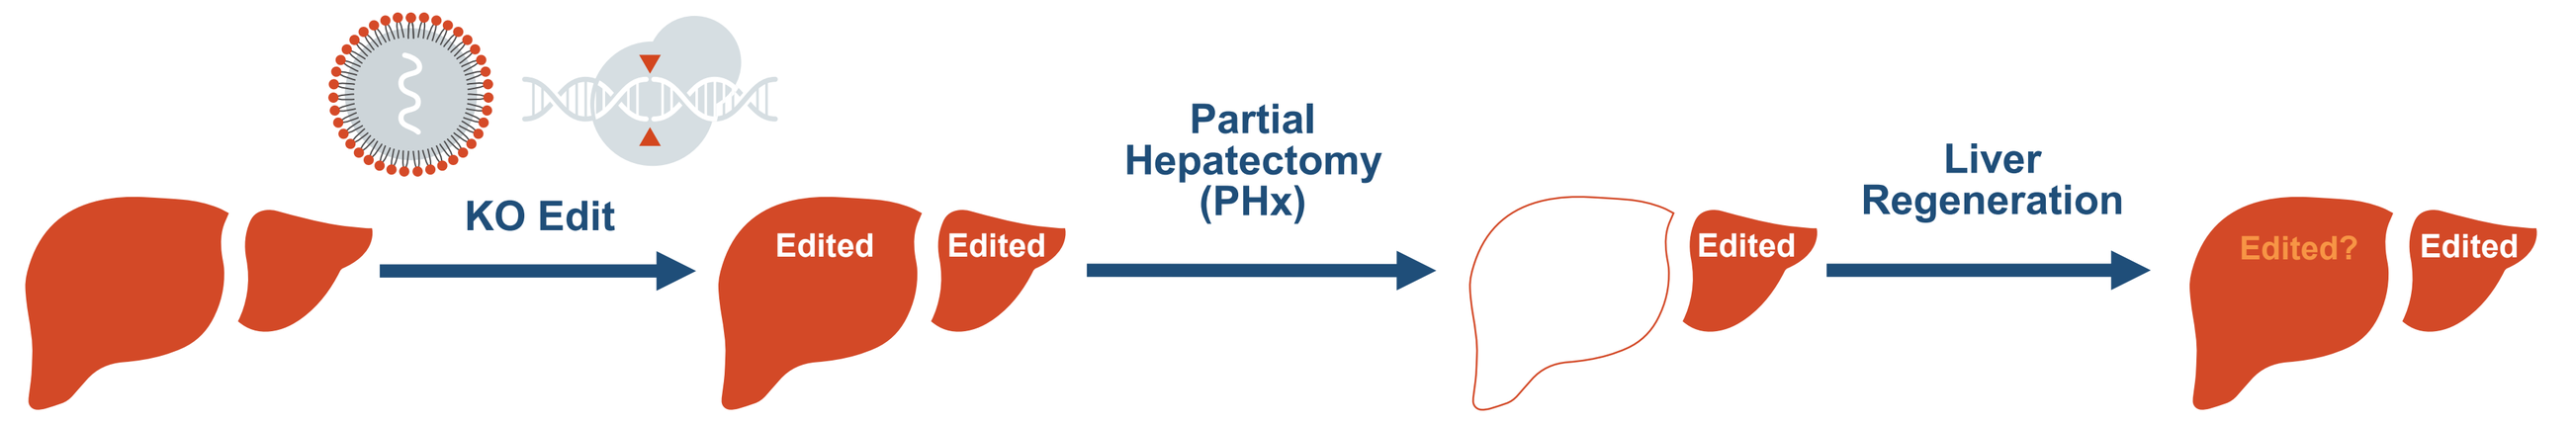
KO Edit
Edited
Edited
Partial Hepatectomy (PHx)
Edited
Liver Regeneration
Edited?
Edited

Methods

Objective: Evaluate durability of liver editing following a 70% partial hepatectomy 14-months after treatment with NTLA-2002 in huKLKB1 mice

- Humanized *KLKB1* mice (huKLKB1) were injected intravenously with NTLA-2002 at 0.1 mg/kg and 0.3 mg/kg or TSS (tris-sucrose-saline), vehicle control.
- After 14-months, a sham or partial hepatectomy surgery (PHx) was performed. Liver tissue and blood was collected during PHx and at necropsy for editing and serum pre-kallikrein analysis, respectively.

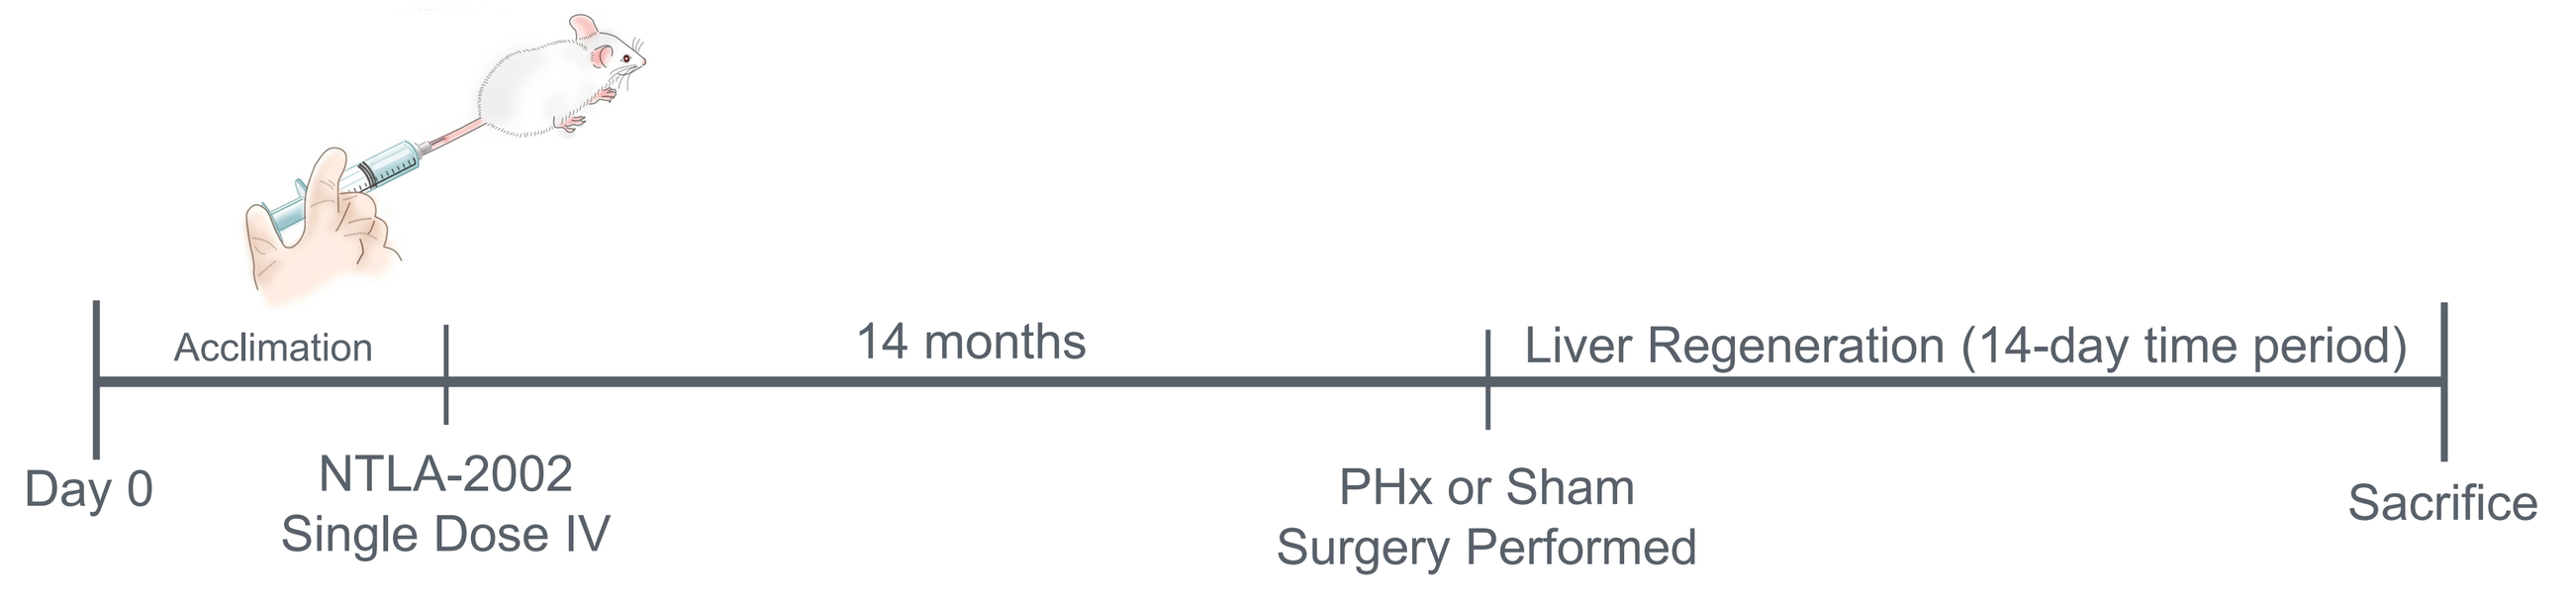
Acclimation
14 months
Liver Regeneration (14-day time period)
Day 0
NTLA-2002
Single Dose IV
PHx or Sham
Surgery Performed
Sacrifice

Durability of Effect with NTLA-2002 in huKLKB1 Mouse Model

Durable and robust liver editing and serum knockdown in huKLKB1 mice 12 months after single dose treatment with NTLA-2002

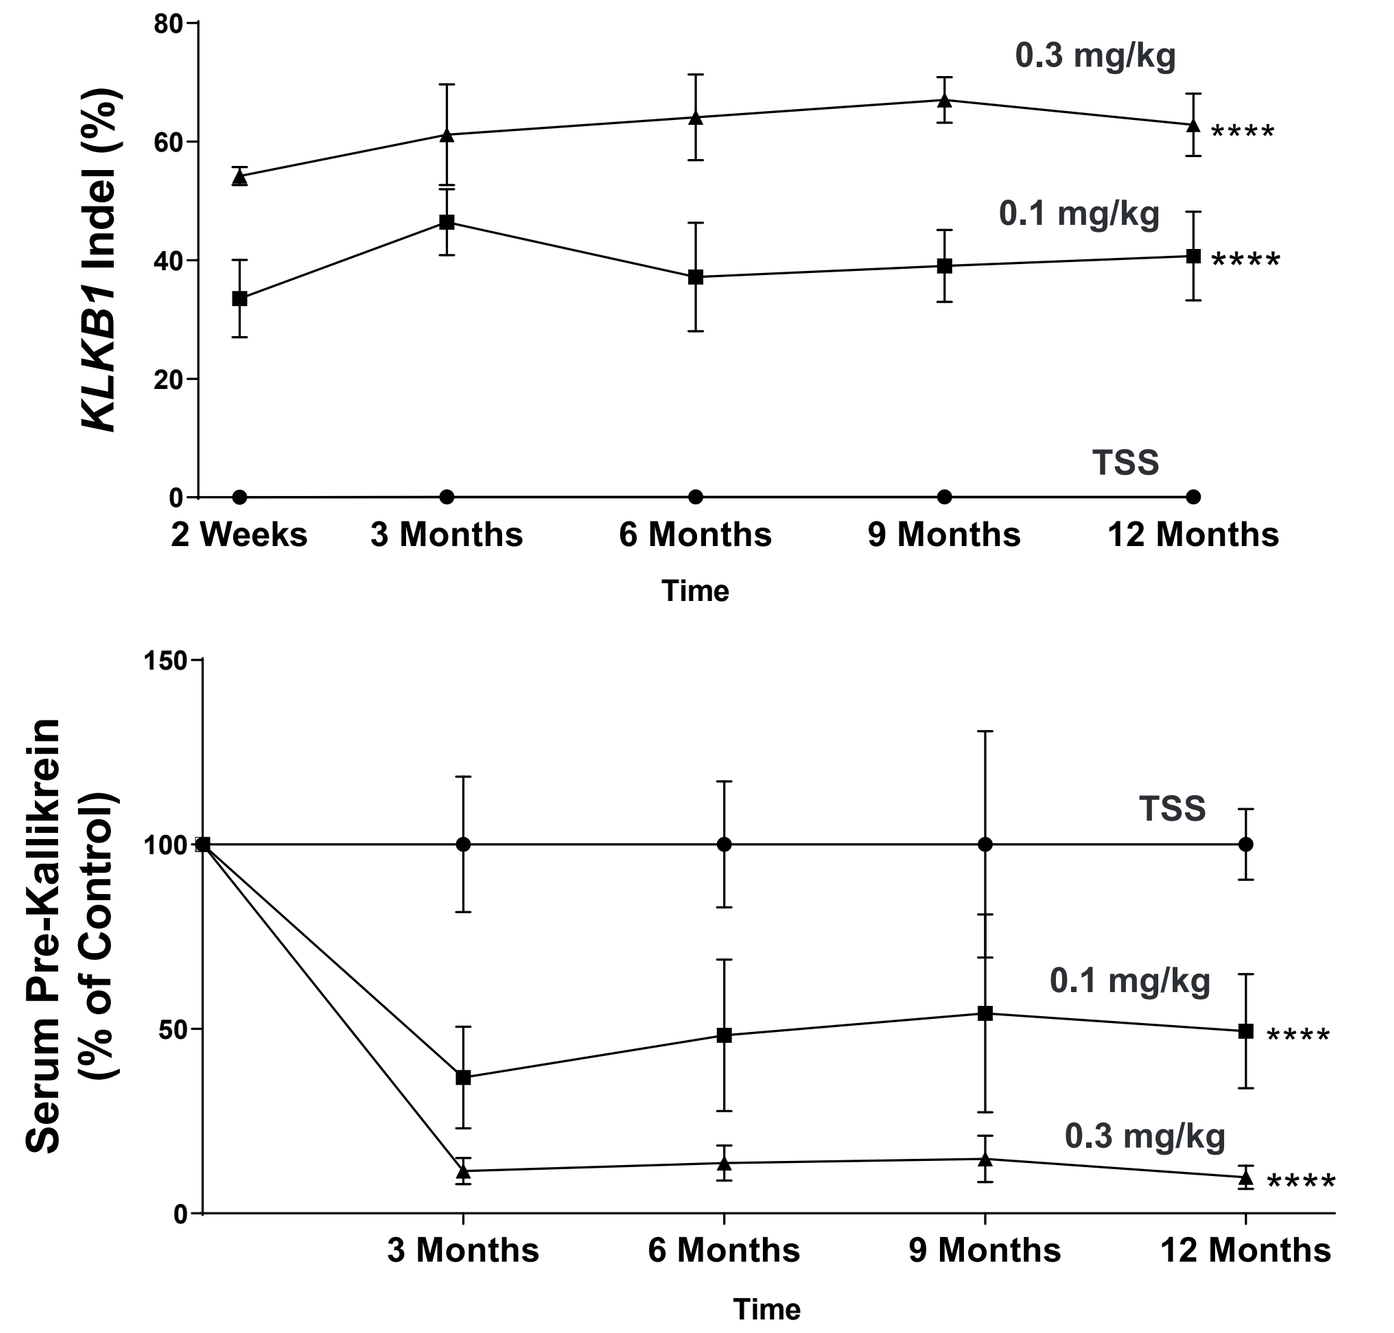
KLKB1 Indel (%)
0.3 mg/kg
0.1 mg/kg
TSS
2 Weeks
3 Months
6 Months
9 Months
12 Months
Time
Serum Pre-Kallikrein (% of Control)
TSS
0.1 mg/kg
0.3 mg/kg
3 Months
6 Months
9 Months
12 Months
Time

Treatment	Dose (mg/kg)	*KLKB1* Indel (%)	Serum Pre-Kallikrein (% of Control)
TSS	N/A	0.09	0.00
NTLA-2002	0.1	40.70	49.37
NTLA-2002	0.3	62.84	9.8

The mean plot of percent *KLKB1* genome editing in liver tissue 12 months post-dose following i.v. administration of 0.3 mg/kg NTLA-2002, 0.1 mg/kg NTLA-2002, or TSS. The mean plot of human serum pre-kallikrein levels expressed as % of Control (TSS) from pre-bleed values at 3 months (m), 6m, 9m and 12m post-dose following i.v. administration of 0.3 mg/kg NTLA-2002, 0.1 mg/kg NTLA-2002, or TSS. Data are mean ± SD (N=8/group). ****p<0.0001 vs. TSS group.

CRISPR/Cas9-mediated gene edits remain unchanged following partial hepatectomy

14 months after NTLA-2002 treatment, editing and serum pre-kallikrein knockdown is maintained in regenerated liver following 70% partial hepatectomy

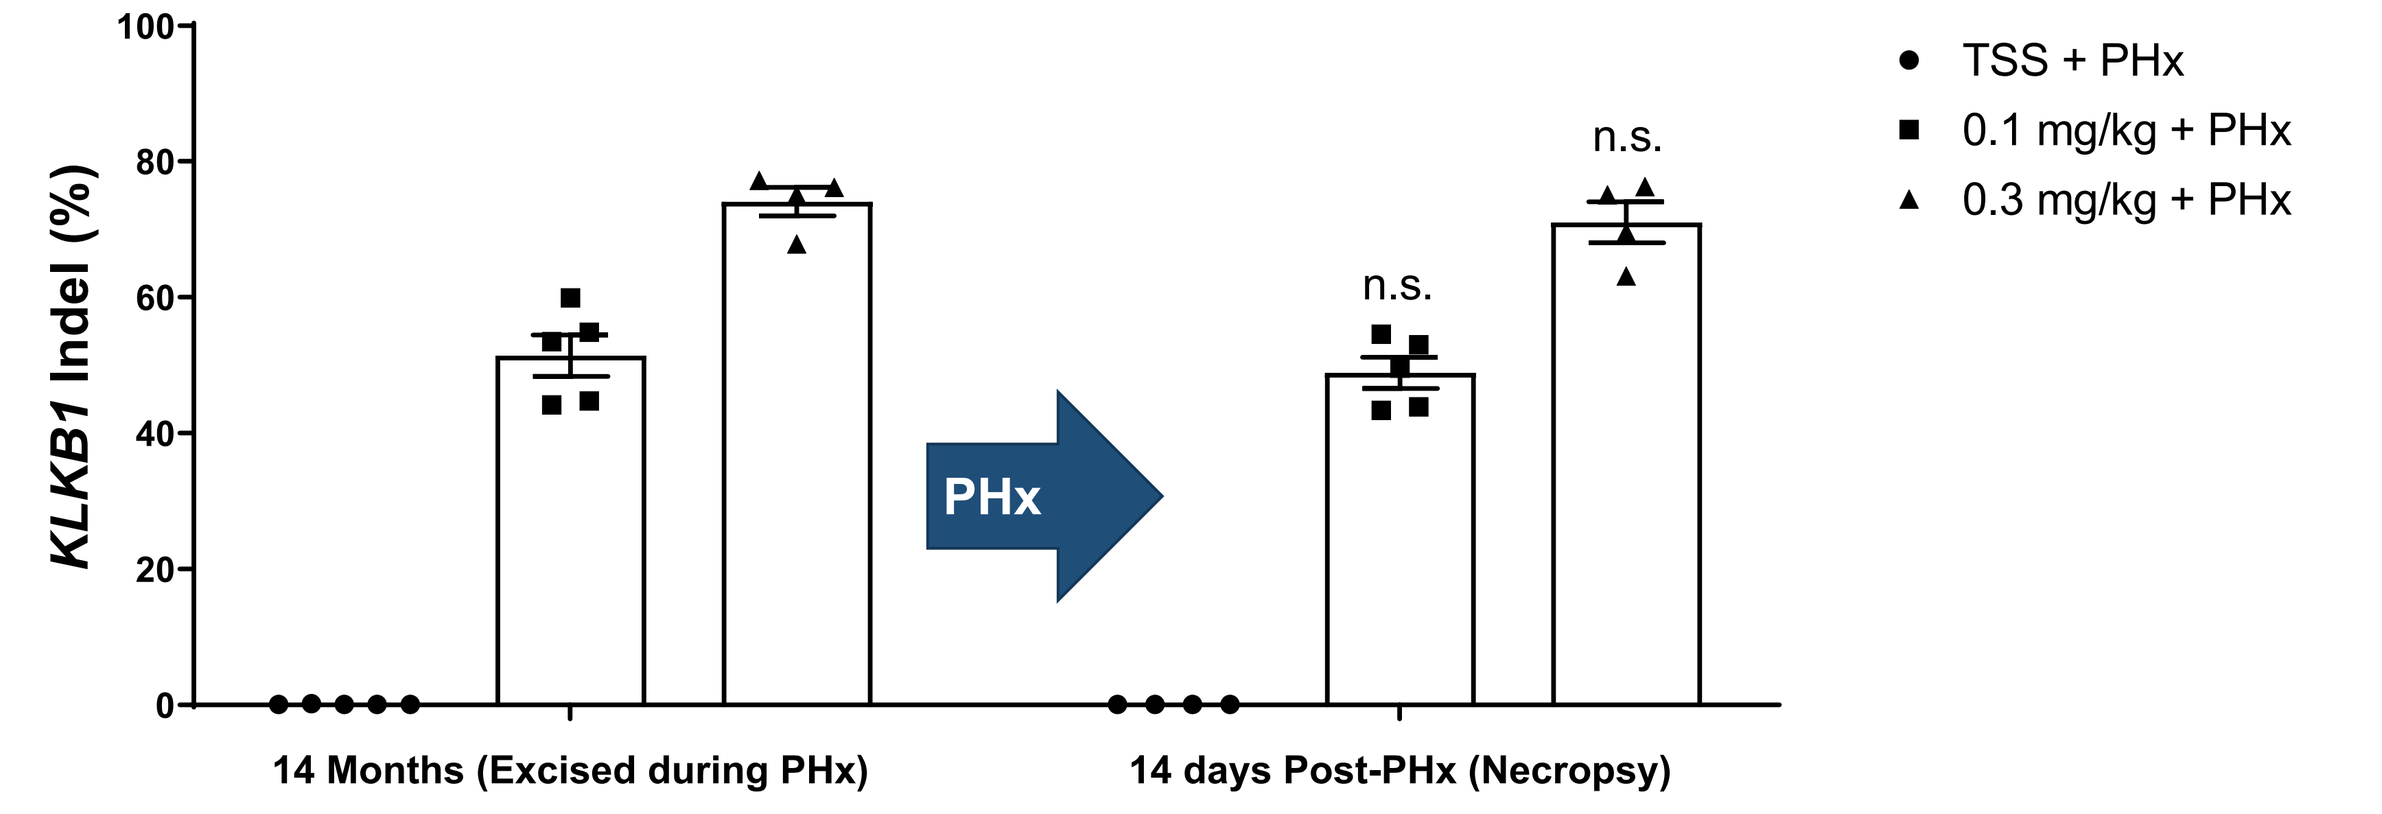
KLKB1 Indel (%)
TSS + PHx
0.1 mg/kg + PHx
0.3 mg/kg + PHx
n.s.
n.s.
PHx
14 Months (Excised during PHx)
14 days Post-PHx (Necropsy)

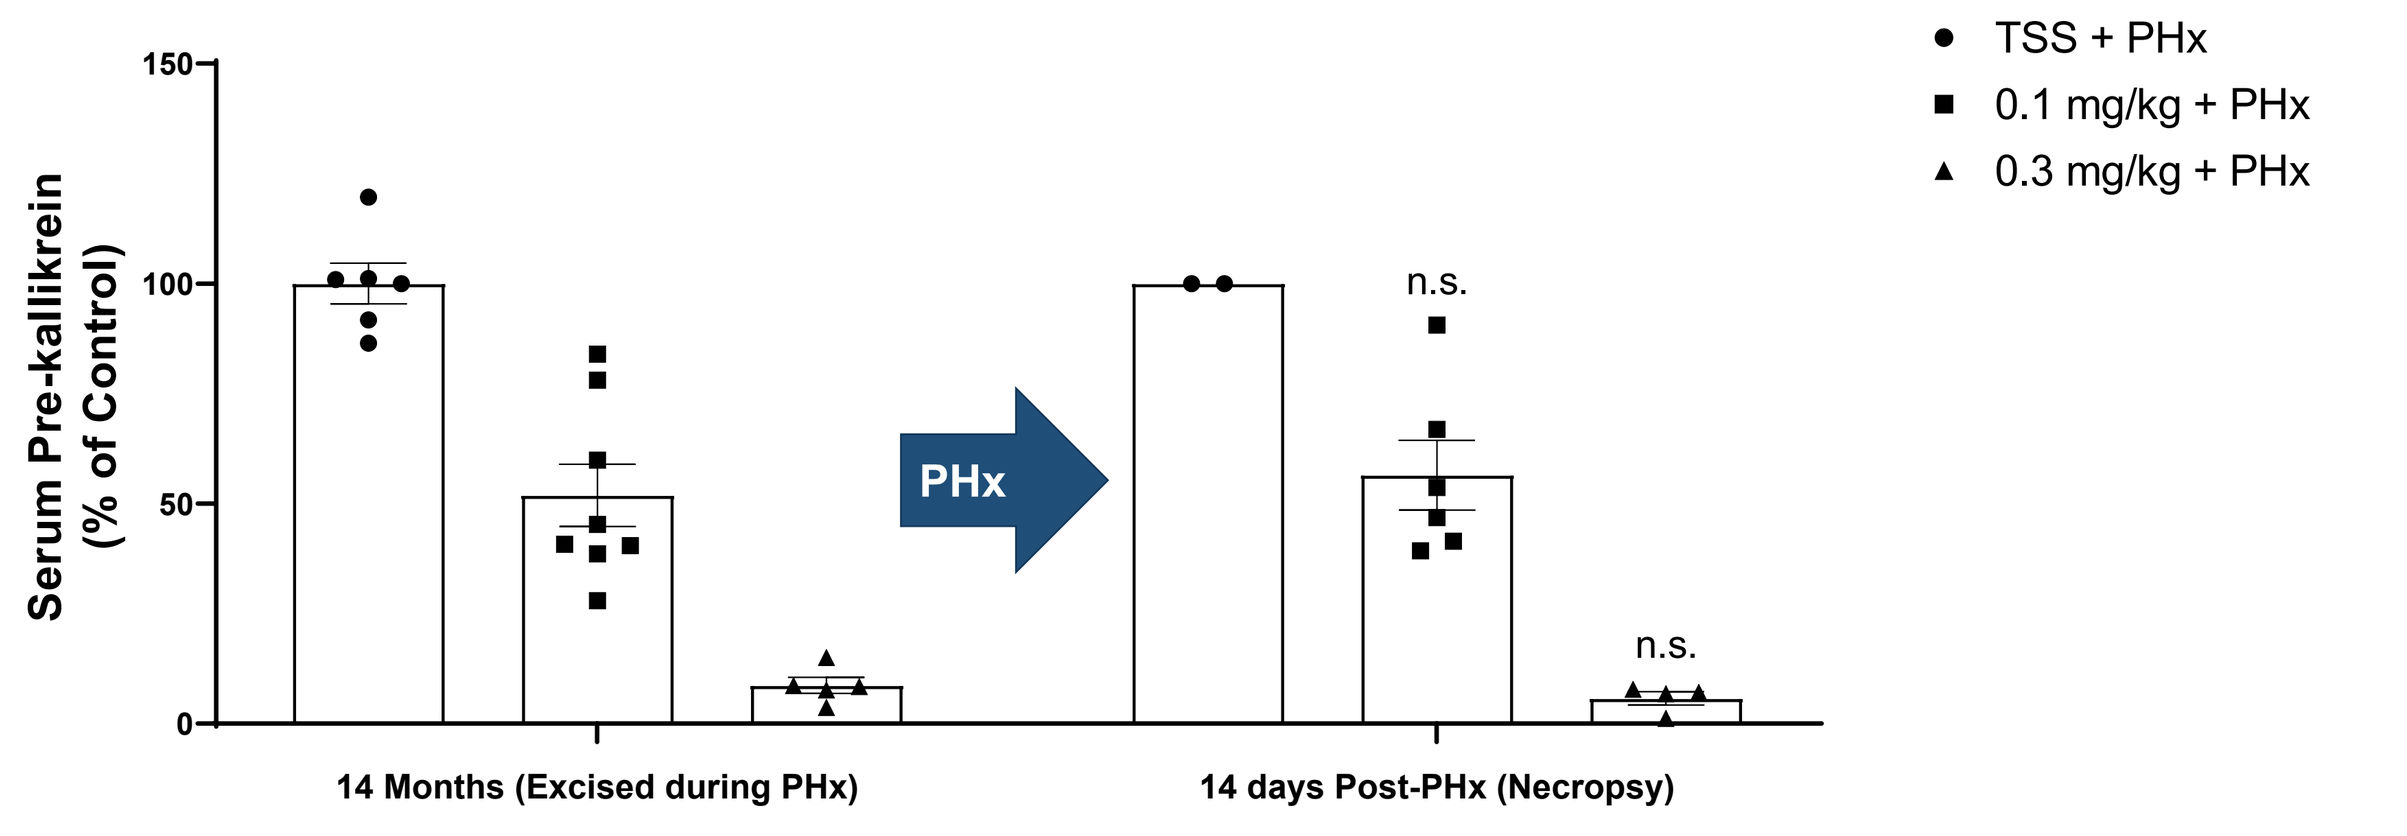
Serum Pre-kallikrein (% of Control)
TSS + PHx
0.1 mg/kg + PHx
0.3 mg/kg + PHx
n.s.
n.s.
PHx
14 Months (Excised during PHx)
14 days Post-PHx (Necropsy)

The mean plot of percent *KLKB1* genome editing in liver tissue taken at time of excision following partial hepatectomy (PHx) and at necropsy 14-days post-operation following i.v. administration of 0.3 mg/kg or 0.1 mg/kg NTLA-2002 or TSS, 14 months prior. The mean plot of The mean plot of human serum pre-kallikrein levels expressed as % of Control (TSS) from 14 months post-dose following i.v administration of 0.3 mg/kg or 0.1 mg/kg NTLA-2002 or TSS and 14 days post-operation (day 14). Data are mean ± SEM (N=5-8/group). **p<0.001 vs. Respective Pre-PHx value. n.s. = not significant

Key Takeaways

- NTLA-2002 is a non-viral, CRISPR/Cas9 therapeutic candidate designed to knock out the hepatic *KLKB1* gene as a single-dose treatment for HAE.
- A single administration of NTLA-2002 shows sustained and durable editing and corresponding serum pre-kallikrein knockdown >12 months in the huKLKB1 mouse.
- *KLKB1* editing is retained following partial hepatectomy-induced liver regeneration, further demonstrating the durability of CRISPR/Cas9-induced gene editing with NTLA-2002.
- These data further support the development of NTLA-2002 as a potential one-time treatment for patients with HAE.
- NTLA-2002 is currently undergoing Phase 1/2 clinical trials.