POTENT, CRISPR-ENGINEERED OFF-THE-SHELF CAR-T AND TCR-T CELLS EVADING HOST T AND NK CELL REJECTION

#P390

Zhang Y, Prodeus A, Jetley U, Tan y, Ranade U, Balwani I, Liu D, Miller I, Zhang B, Dutta I, Sharma P, Goel S, Malik V, Venkatesan P, Keenan M, Lam M, Arredouani S, **Schultes BC**, Intellia Therapeutics, Inc., Cambridge, MA, USA

INTELLIA'S ALLOGENEIC SOLUTION

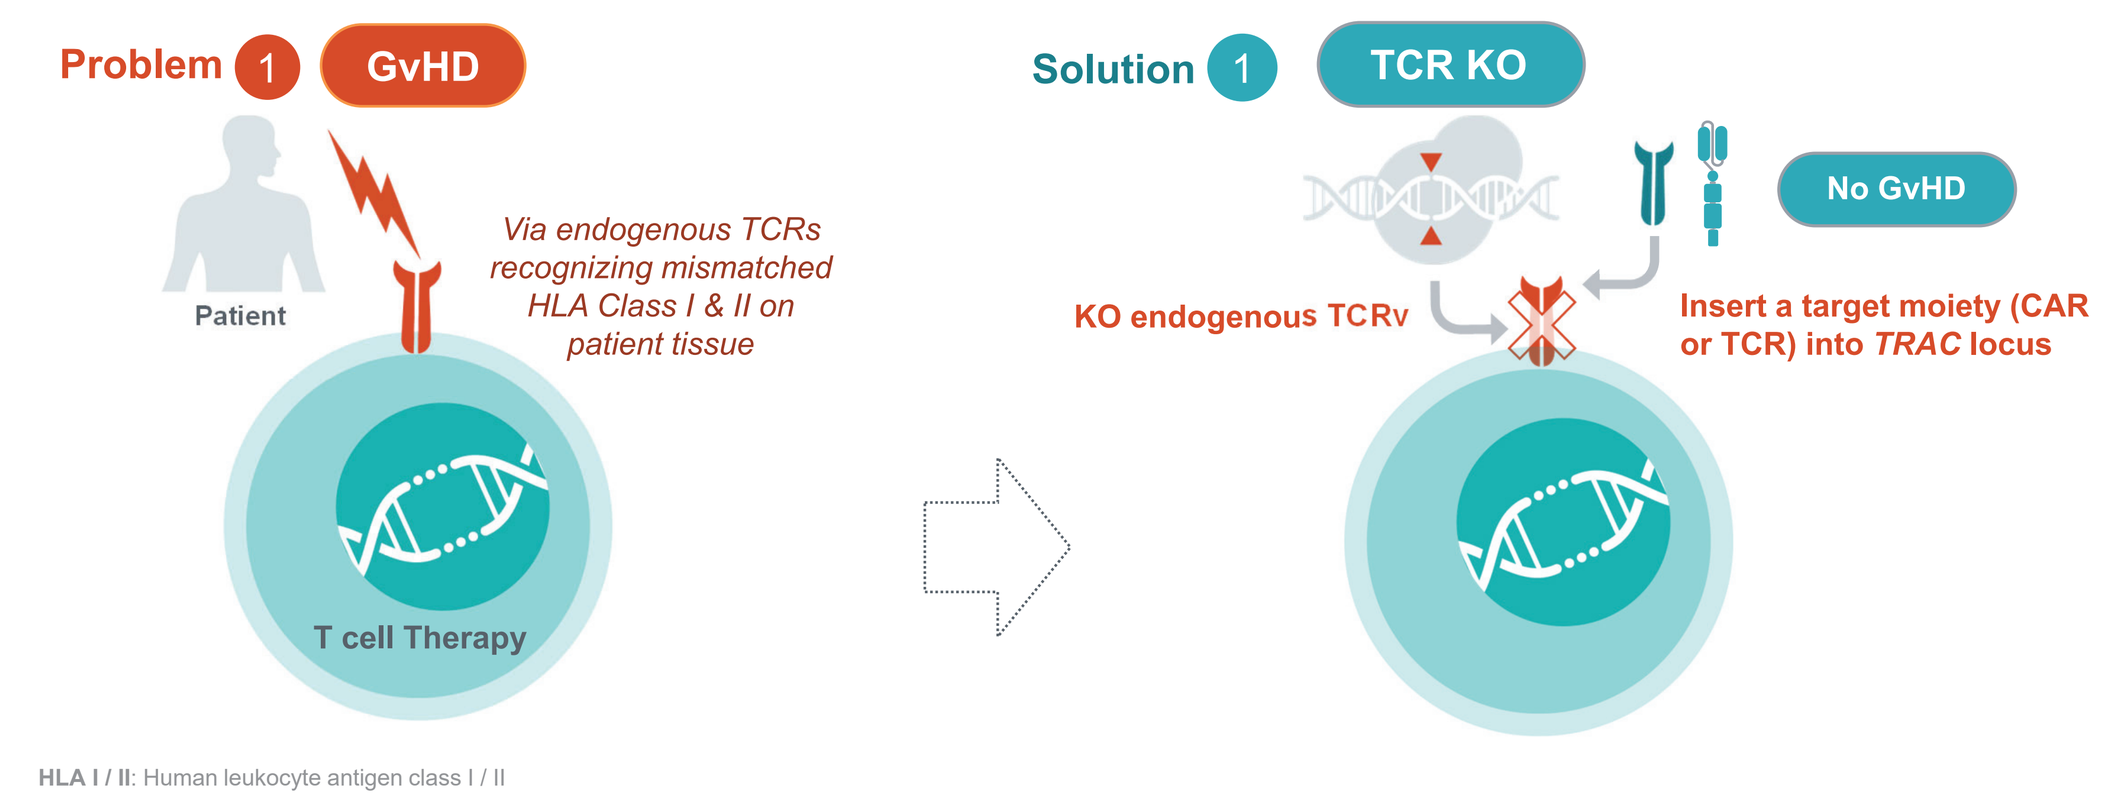
Problem 1 GvHD
Patient
Via endogenous TCRs recognizing mismatched HLA Class I & II on patient tissue
T cell Therapy
Solution 1 TCR KO
No GvHD
KO endogenous TCRv
Insert a target moiety (CAR or TCR) into TRAC locus
HLA I / II: Human leukocyte antigen class I / II

1 Graft versus host disease (GvHD)
T cell receptor (TCR) from allogeneic T cells recognizes and kills recipient (host) cells.
Primary safety concern

✓ **Solved** with knockout (KO) of endogenous TCR
✓ Site-specific integration of CAR or tgTCR minimizes insertional mutagenesis risk

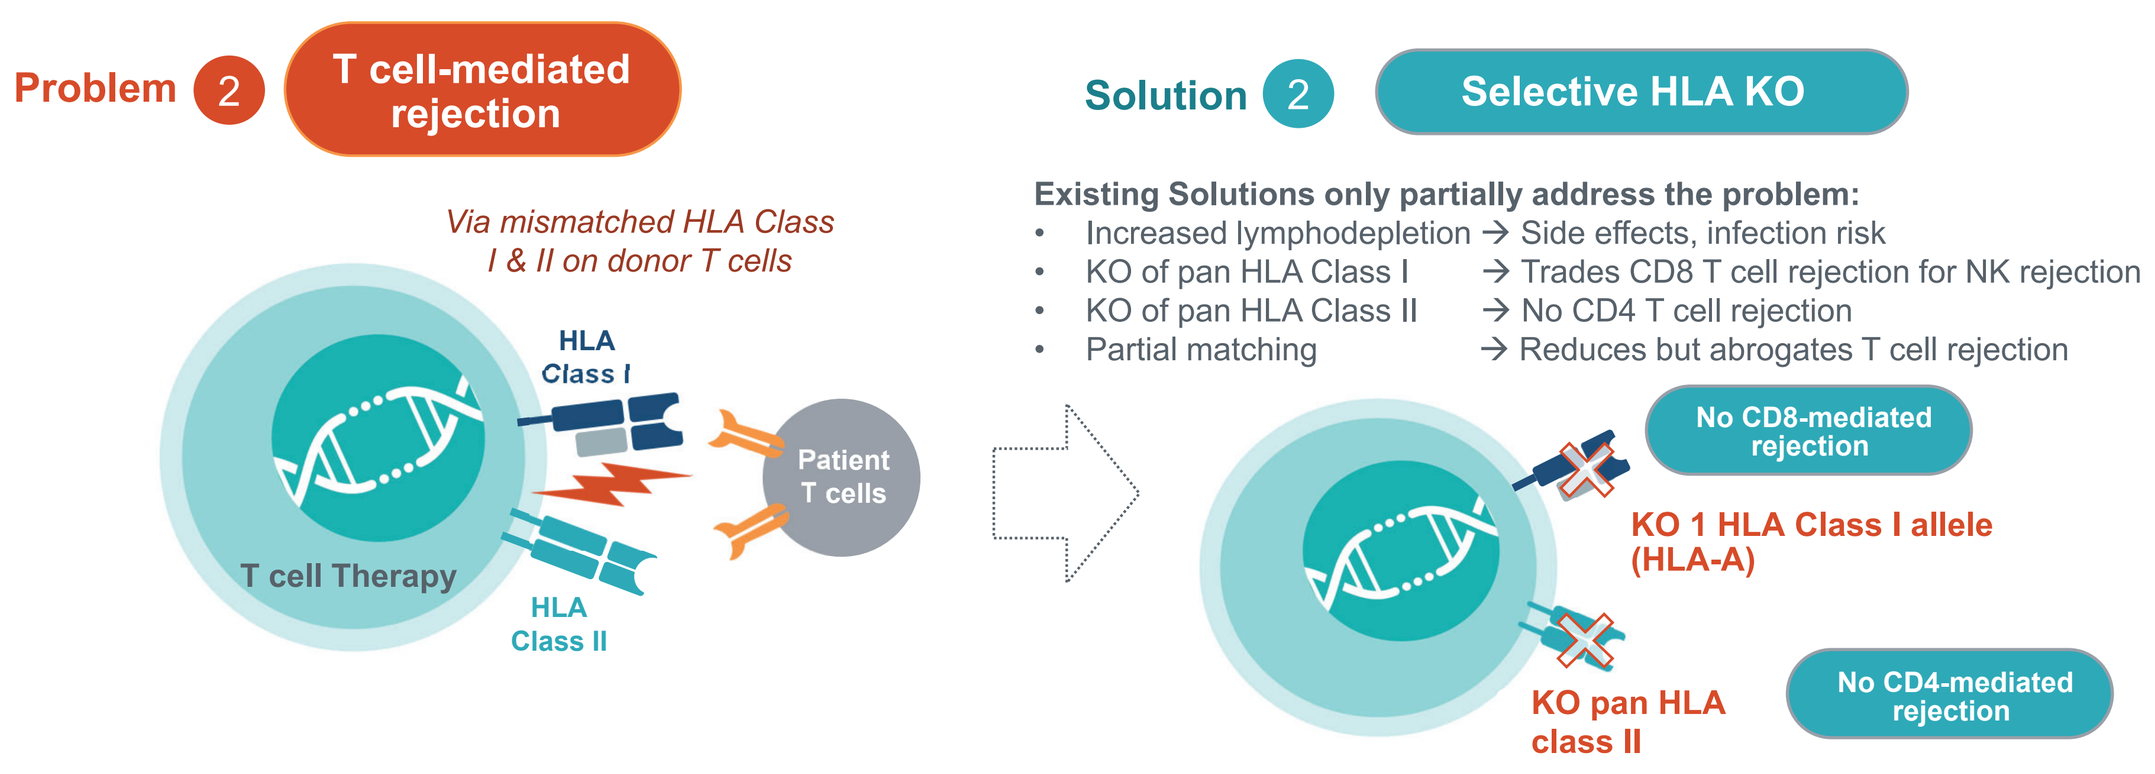
Problem 2 T cell-mediated rejection
Via mismatched HLA Class I & II on donor T cells
HLA Class I
Patient T cells
T cell Therapy
HLA Class II
Solution 2 Selective HLA KO
No CD8-mediated rejection
KO 1 HLA Class I allele (HLA-A)
KO pan HLA class II
No CD4-mediated rejection

Existing Solutions only partially address the problem:
- Increased lymphodepletion → Side effects, infection risk
- KO of pan HLA Class I → Trades CD8 T cell rejection for NK rejection
- KO of pan HLA Class II → No CD4 T cell rejection
- Partial matching → Reduces but abrogates T cell rejection

2 Rejection via host T cells
Human Leukocyte Antigen (HLA) molecules must match between donor and recipient to prevent rejection from:
- Host CD8 T cells via HLA Class I
- Host CD4 T cells via HLA Class II

Primary persistence concern

Solved with:
✓ KO of HLA Class II
✓ KO of HLA-A
✓ Match remaining HLA class I alleles
= No mis-matched HLA

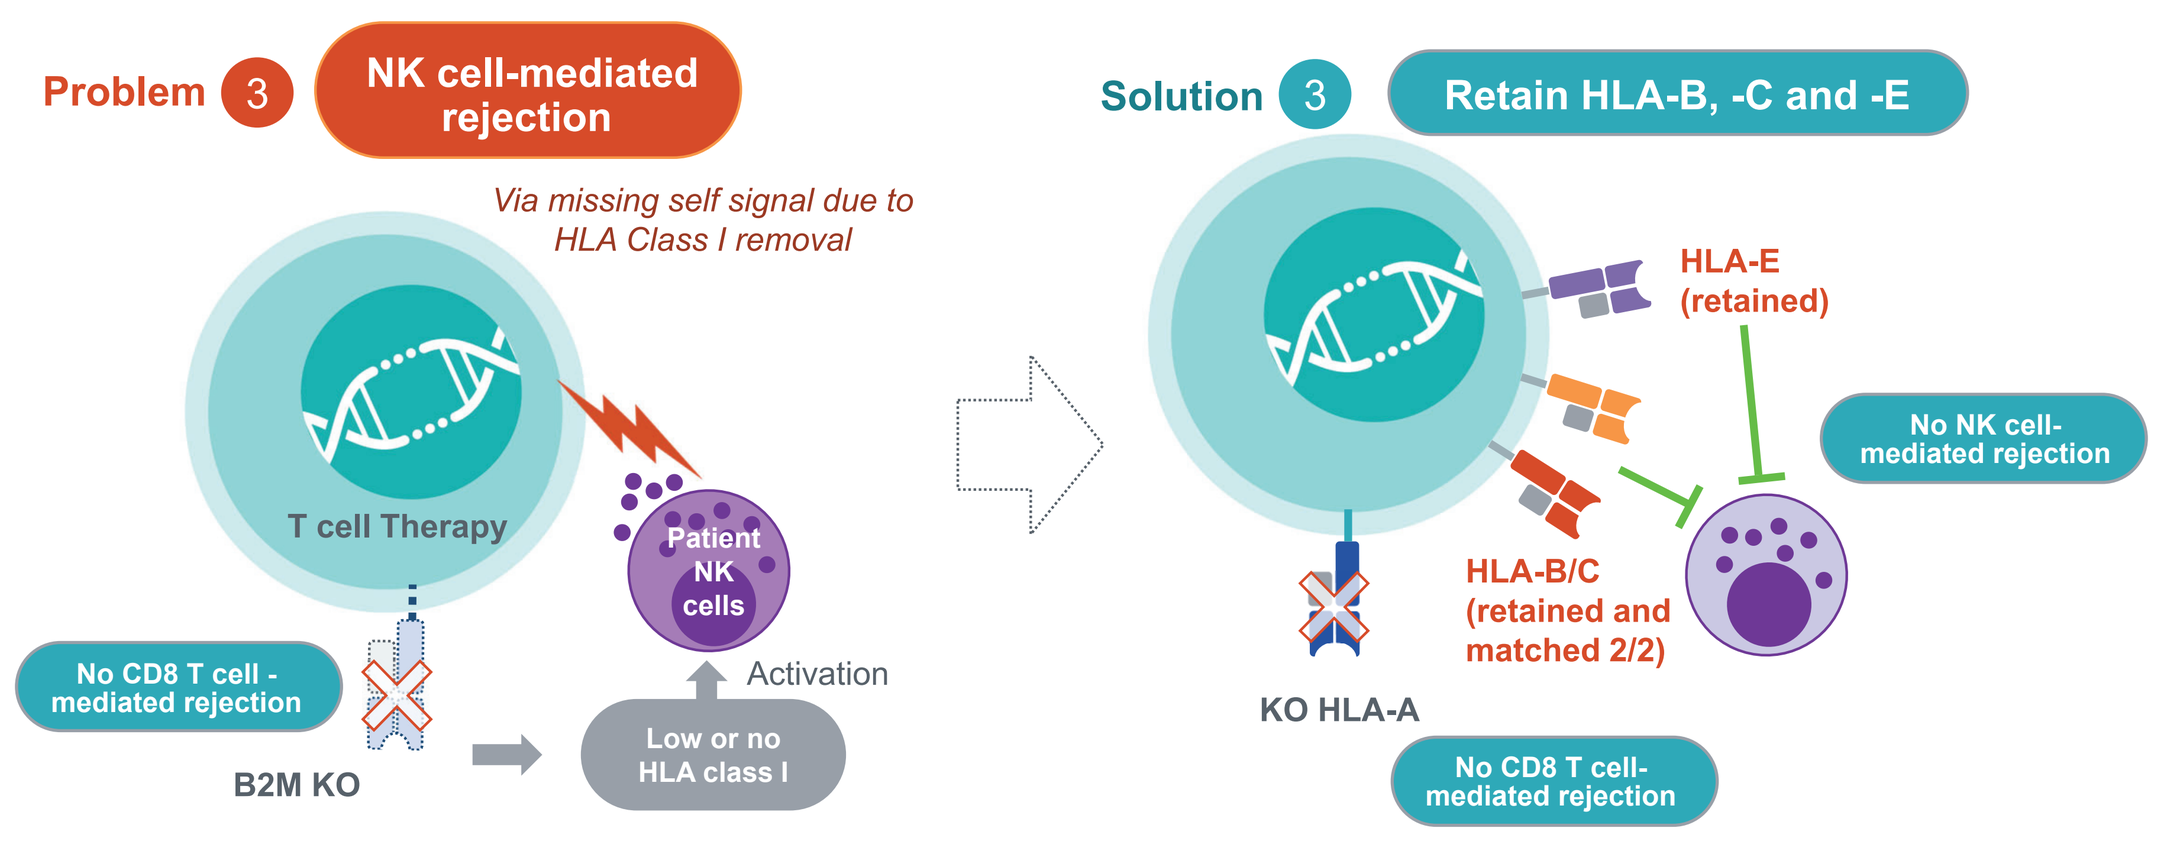
Problem 3 NK cell-mediated rejection
Via missing self signal due to HLA Class I removal
T cell Therapy
Patient NK cells
Activation
No CD8 T cell - mediated rejection
Low or no HLA class I
B2M KO
Solution 3 Retain HLA-B, -C and -E
HLA-E (retained)
No NK cell-mediated rejection
HLA-B/C (retained and matched 2/2)
KO HLA-A
No CD8 T cell-mediated rejection

3 Rejection via host natural killer (NK) cells
NK cells will attack cells that lack HLA-I expression or have low HLA-I
Induced persistence concern

Protection from host NK cells
- NK cell inhibitory receptors engage retained HLA-B, HLA-C, and HLA-E, blocking NK cell activation

The Advantage of a Simplified HLA Matching Strategy:

A Manageable Number of Off-the-Shelf Product Lots Can Be Fully Matched to a Broad Patient Population

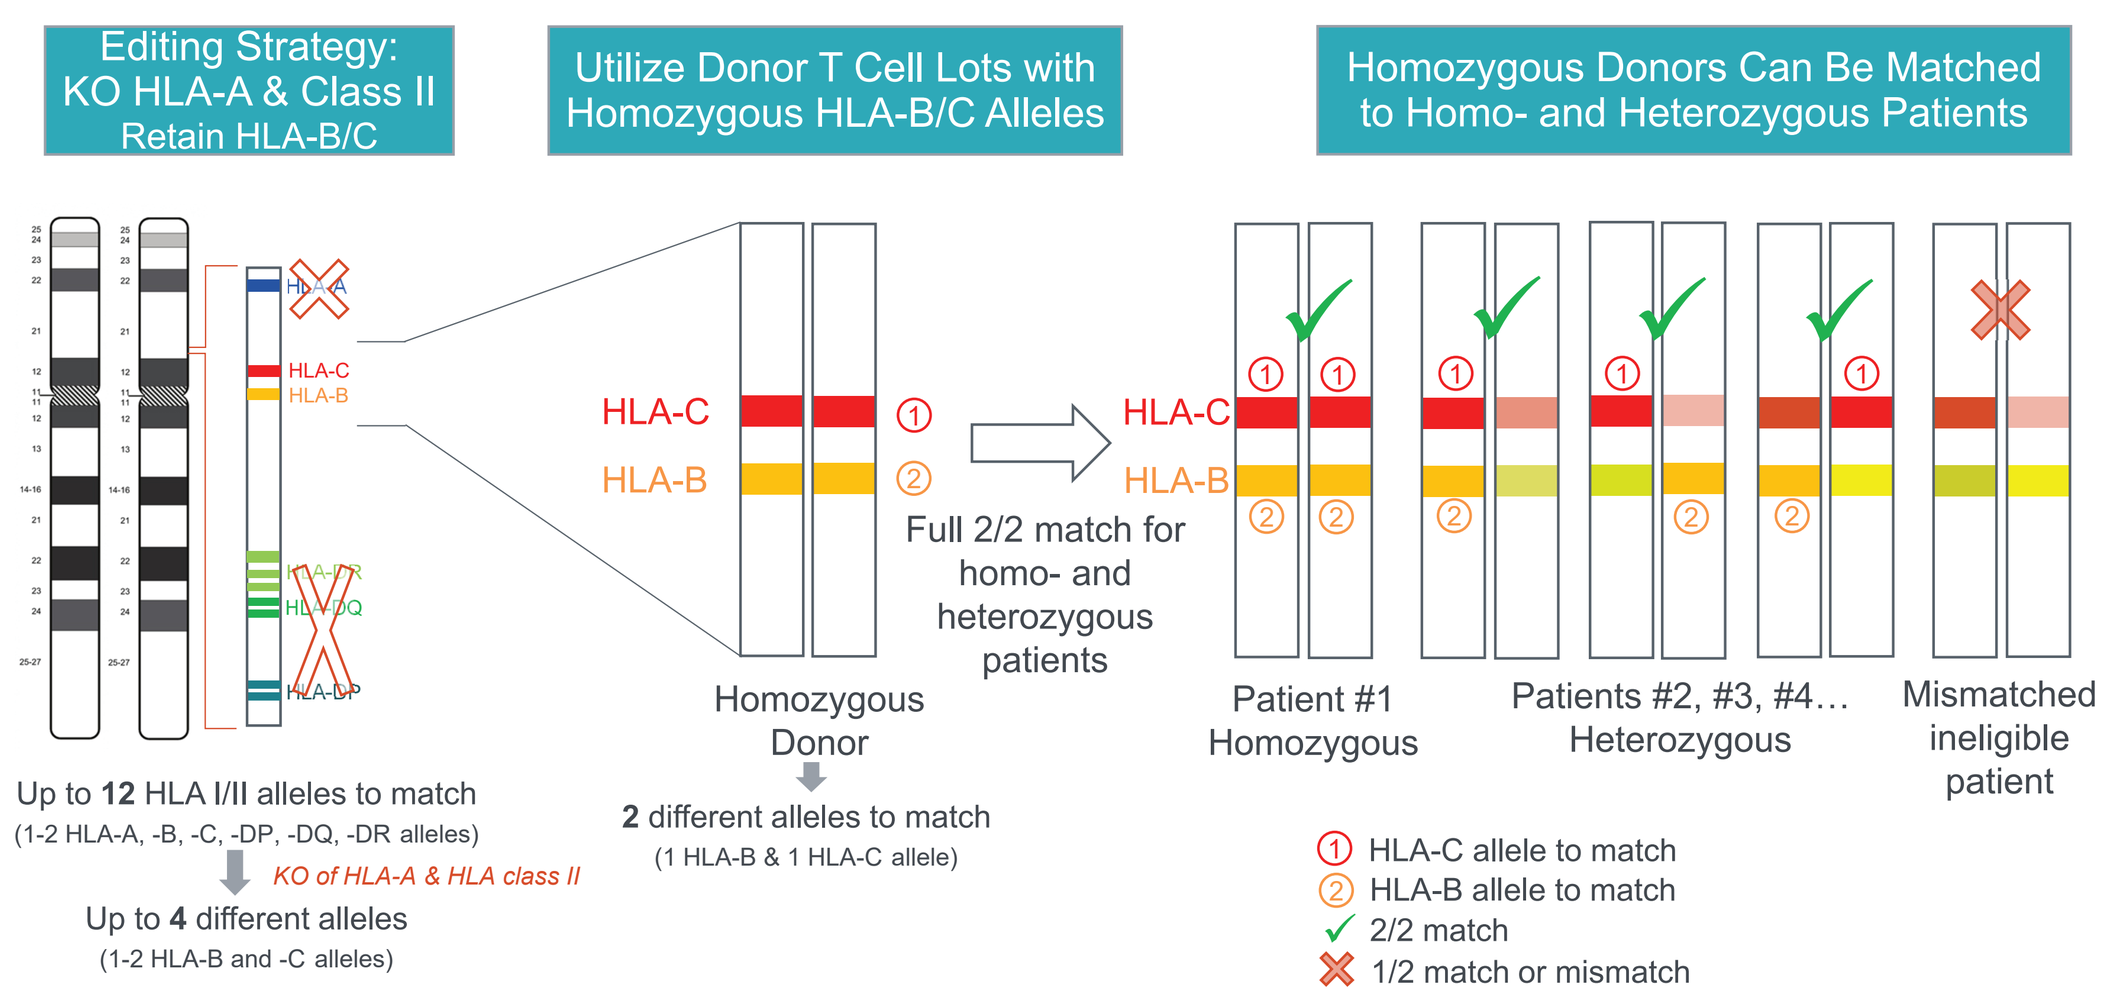
Editing Strategy: KO HLA-A & Class II Retain HLA-B/C
Utilize Donor T Cell Lots with Homozygous HLA-B/C Alleles
Homozygous Donors Can Be Matched to Homo- and Heterozygous Patients
HLA-C
HLA-B
Full 2/2 match for homo- and heterozygous patients
Homozygous Donor
Patient #1 Homozygous
Patients #2, #3, #4… Heterozygous
Mismatched ineligible patient
Up to 12 HLA I/II alleles to match (1-2 HLA-A, -B, -C, -DP, -DQ, -DR alleles)
KO of HLA-A & HLA class II
Up to 4 different alleles (1-2 HLA-B and -C alleles)
2 different alleles to match (1 HLA-B & 1 HLA-C allele)
① HLA-C allele to match
② HLA-B allele to match
✓ 2/2 match
✗ 1/2 match or mismatch

Bioinformatics Analysis Suggests A Limited Number of Donors (<20) Will Provide Coverage for the Majority of Patients

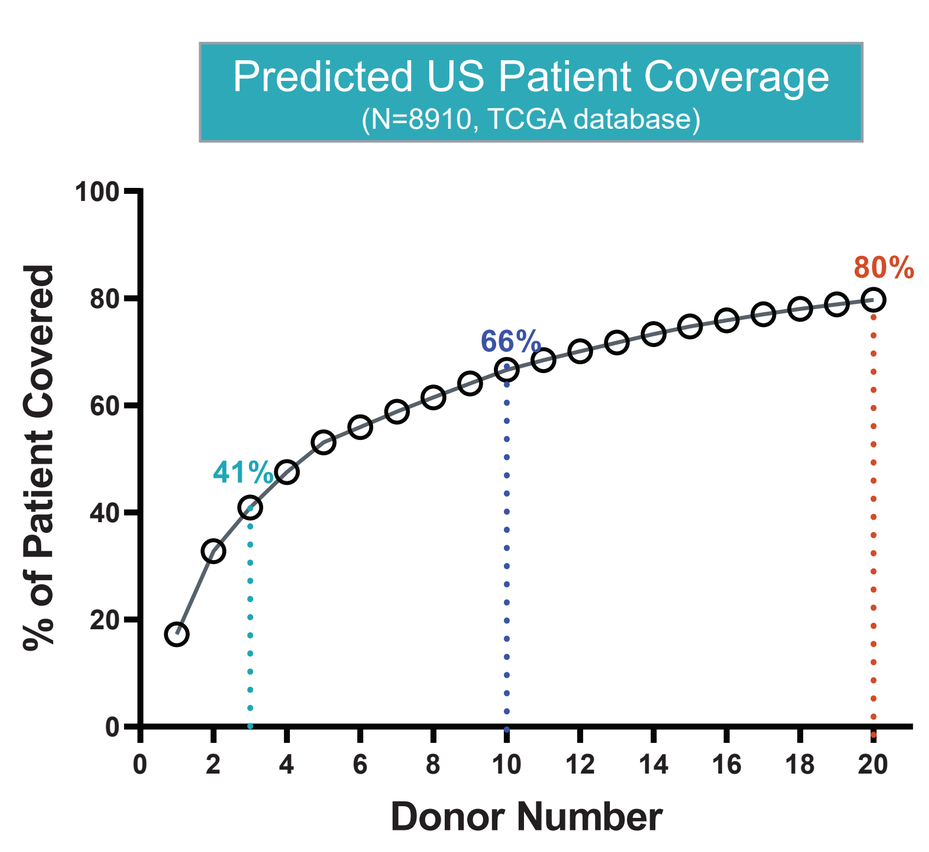
Predicted US Patient Coverage
(N=8910, TCGA database)
% of Patient Covered
Donor Number
41%
66%
80%

- 10 donors cover 66% of the US population
- 20 donors cover 80% of the US population

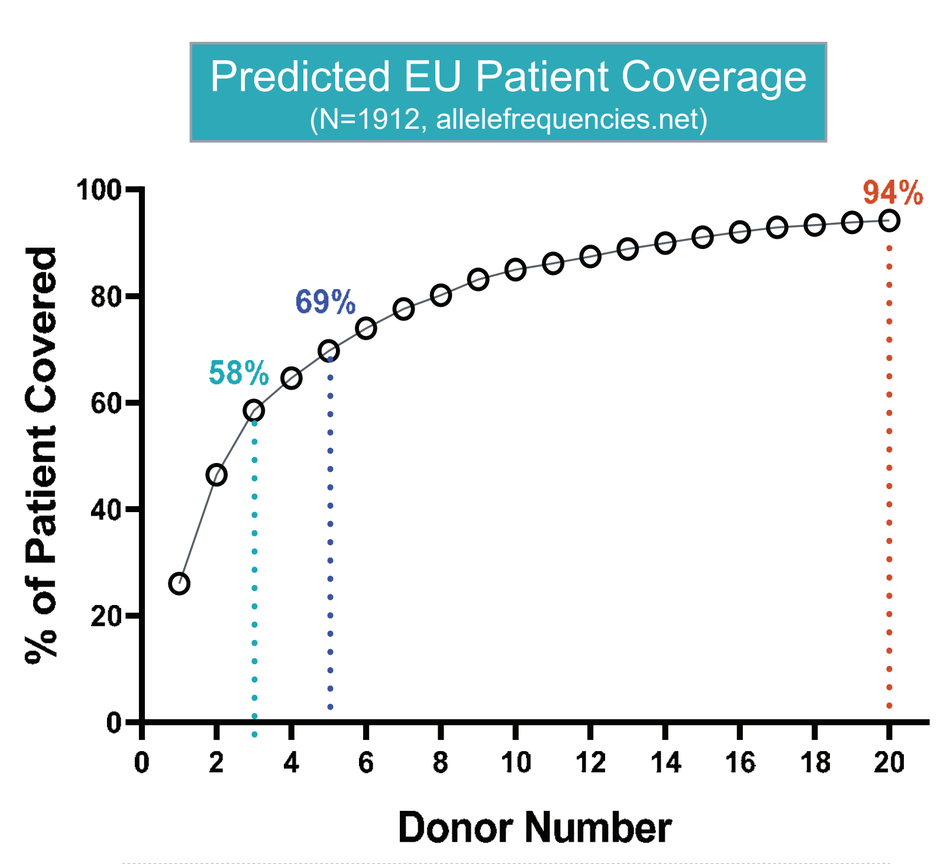
Predicted EU Patient Coverage
(N=1912, allelefrequencies.net)
% of Patient Covered
Donor Number
58%
69%
94%

- 5 donors cover 69% of the EU population
- 20 donors cover 94% of the EU population

Patient coverage estimated based on bioinformatic analyses of large data sets containing individual HLA class I allele information

ADDRESSING GVHD

Mice Engrafted with Allo TCR-KO T Cells Did Not Develop GvHD *in vivo*

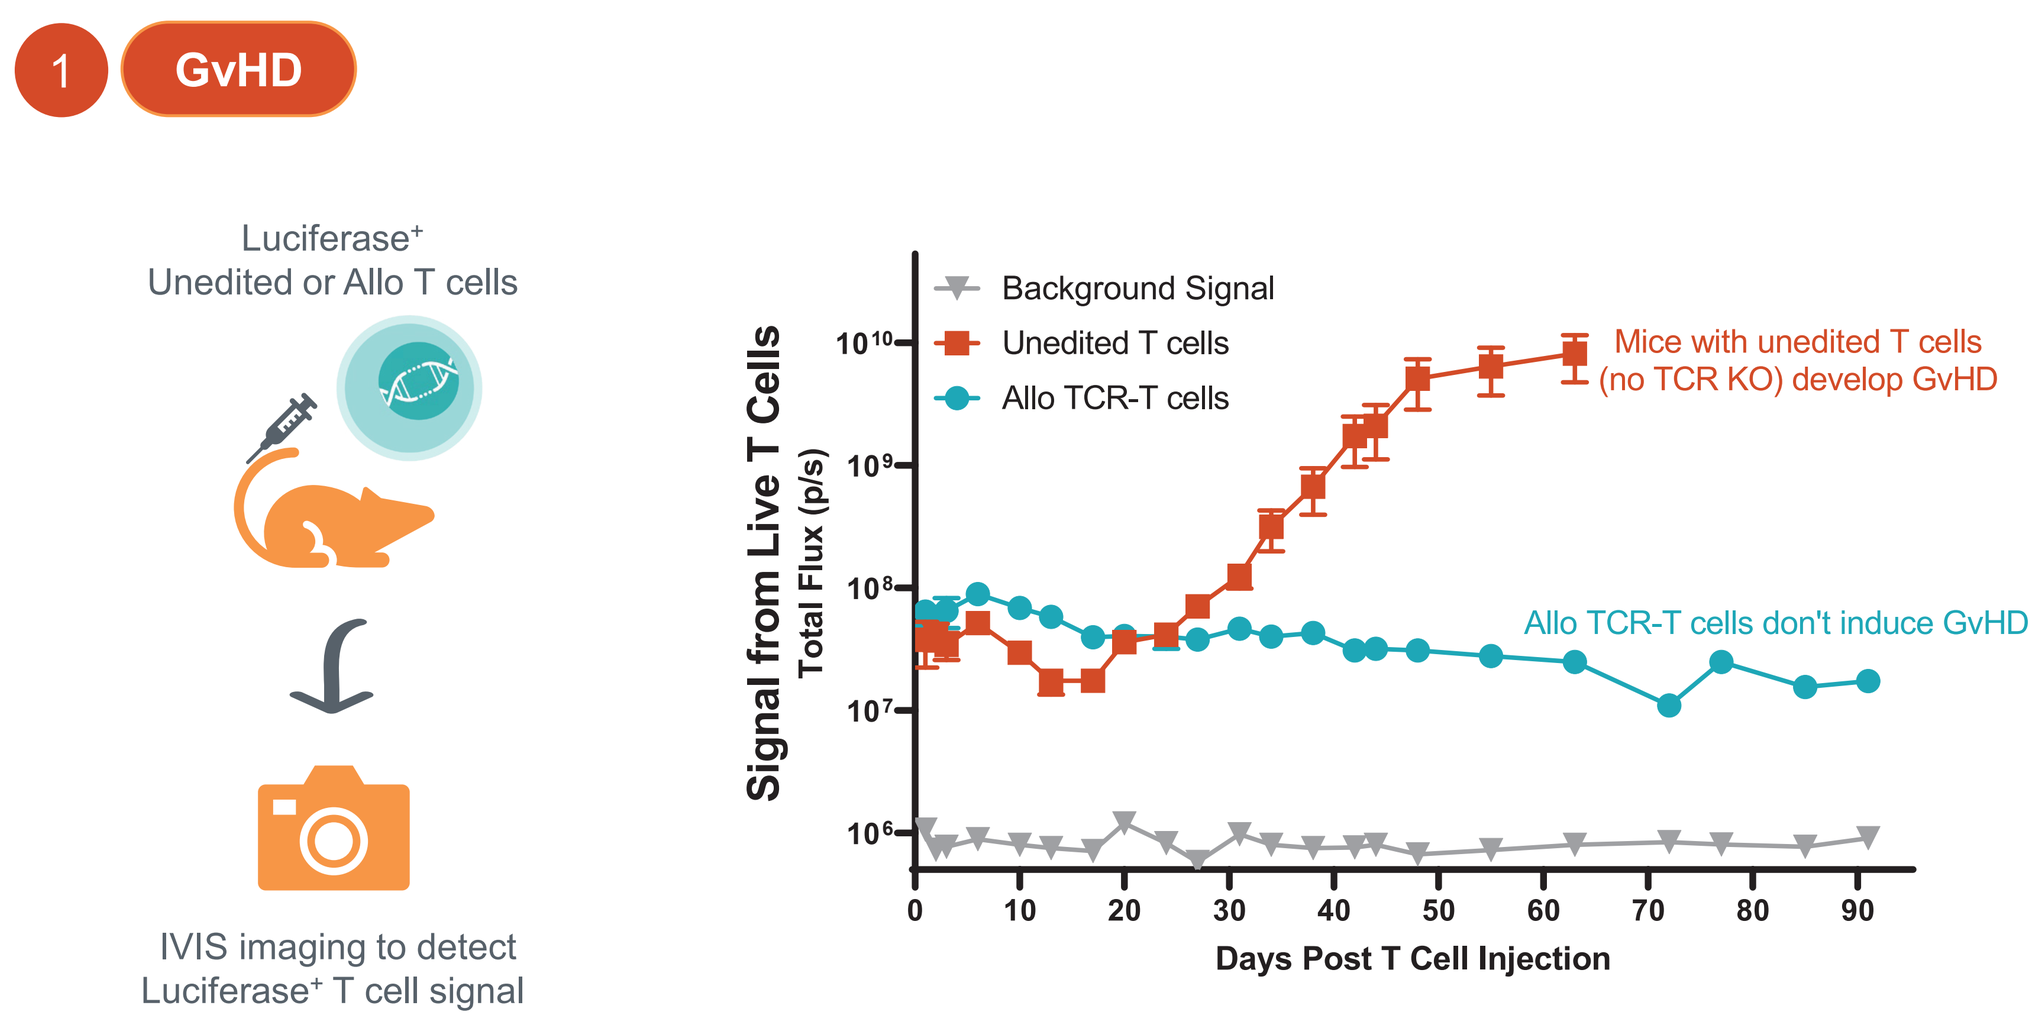
1 GvHD
Luciferase+ Unedited or Allo T cells
IVIS imaging to detect Luciferase+ T cell signal
Background Signal
Unedited T cells
Allo TCR-T cells
Mice with unedited T cells (no TCR KO) develop GvHD
Allo TCR-T cells don't induce GvHD
Signal from Live T Cells
Total Flux (p/s)
Days Post T Cell Injection

ADDRESSING T CELL-MEDIATED REJECTION

Allo T Cells Did Not Elicit Host T Cell Responses *in vitro*

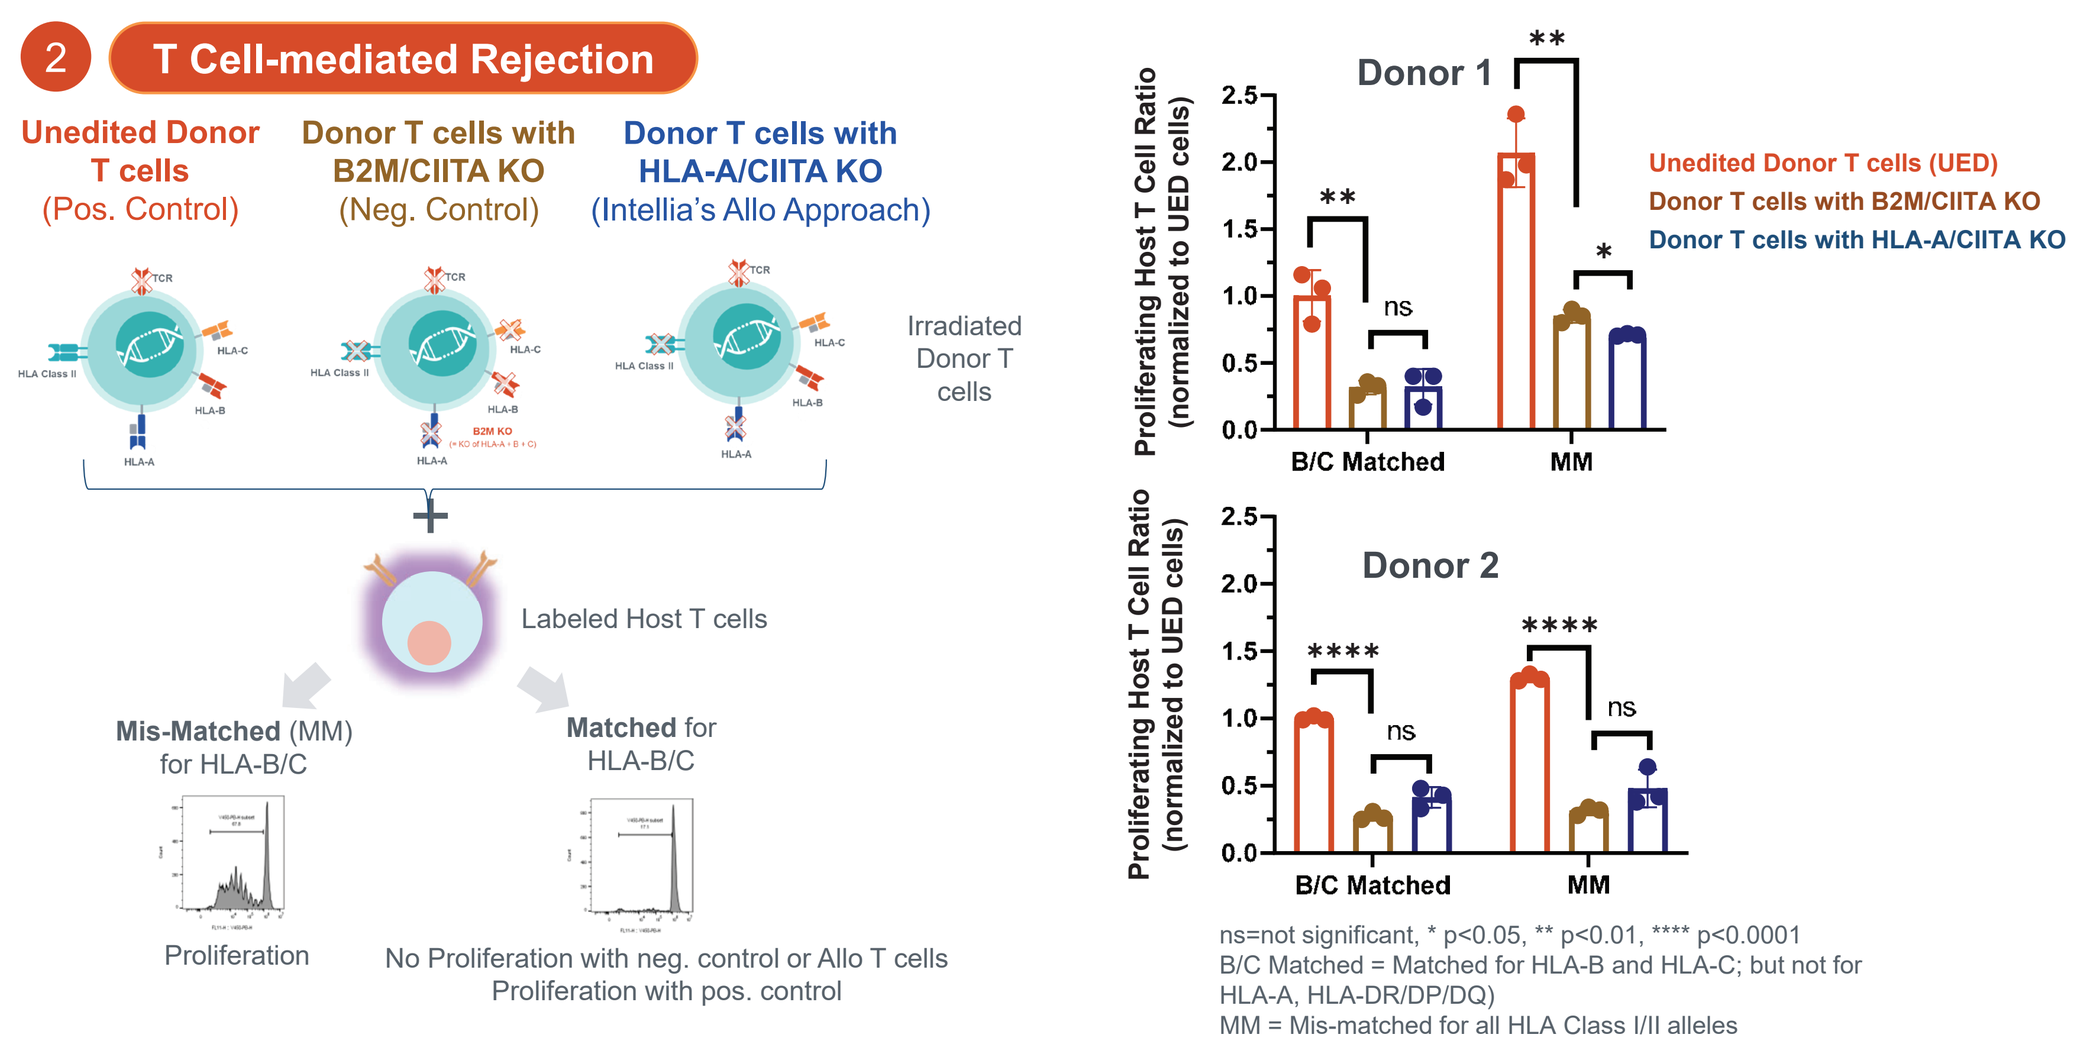
2 T Cell-mediated Rejection
Unedited Donor T cells (Pos. Control)
Donor T cells with B2M/CIITA KO (Neg. Control)
Donor T cells with HLA-A/CIITA KO (Intellia's Allo Approach)
Irradiated Donor T cells
Labeled Host T cells
Mis-Matched (MM) for HLA-B/C
Matched for HLA-B/C
Proliferation
No Proliferation with neg. control or Allo T cells
Proliferation with pos. control
Donor 1
Donor 2
Proliferating Host T Cell Ratio (normalized to UED cells)
B/C Matched
MM
Unedited Donor T cells (UED)
Donor T cells with B2M/CIITA KO
Donor T cells with HLA-A/CIITA KO
ns=not significant, * p<0.05, ** p<0.01, **** p<0.0001
B/C Matched = Matched for HLA-B and HLA-C; but not for HLA-A, HLA-DR/DP/DQ)
MM = Mis-matched for all HLA Class I/II alleles

ADDRESSING HOST NK CELL MEDIATED REJECTION

Intellia's Allo T Cells Were Protected from NK Cell Killing *in vitro*

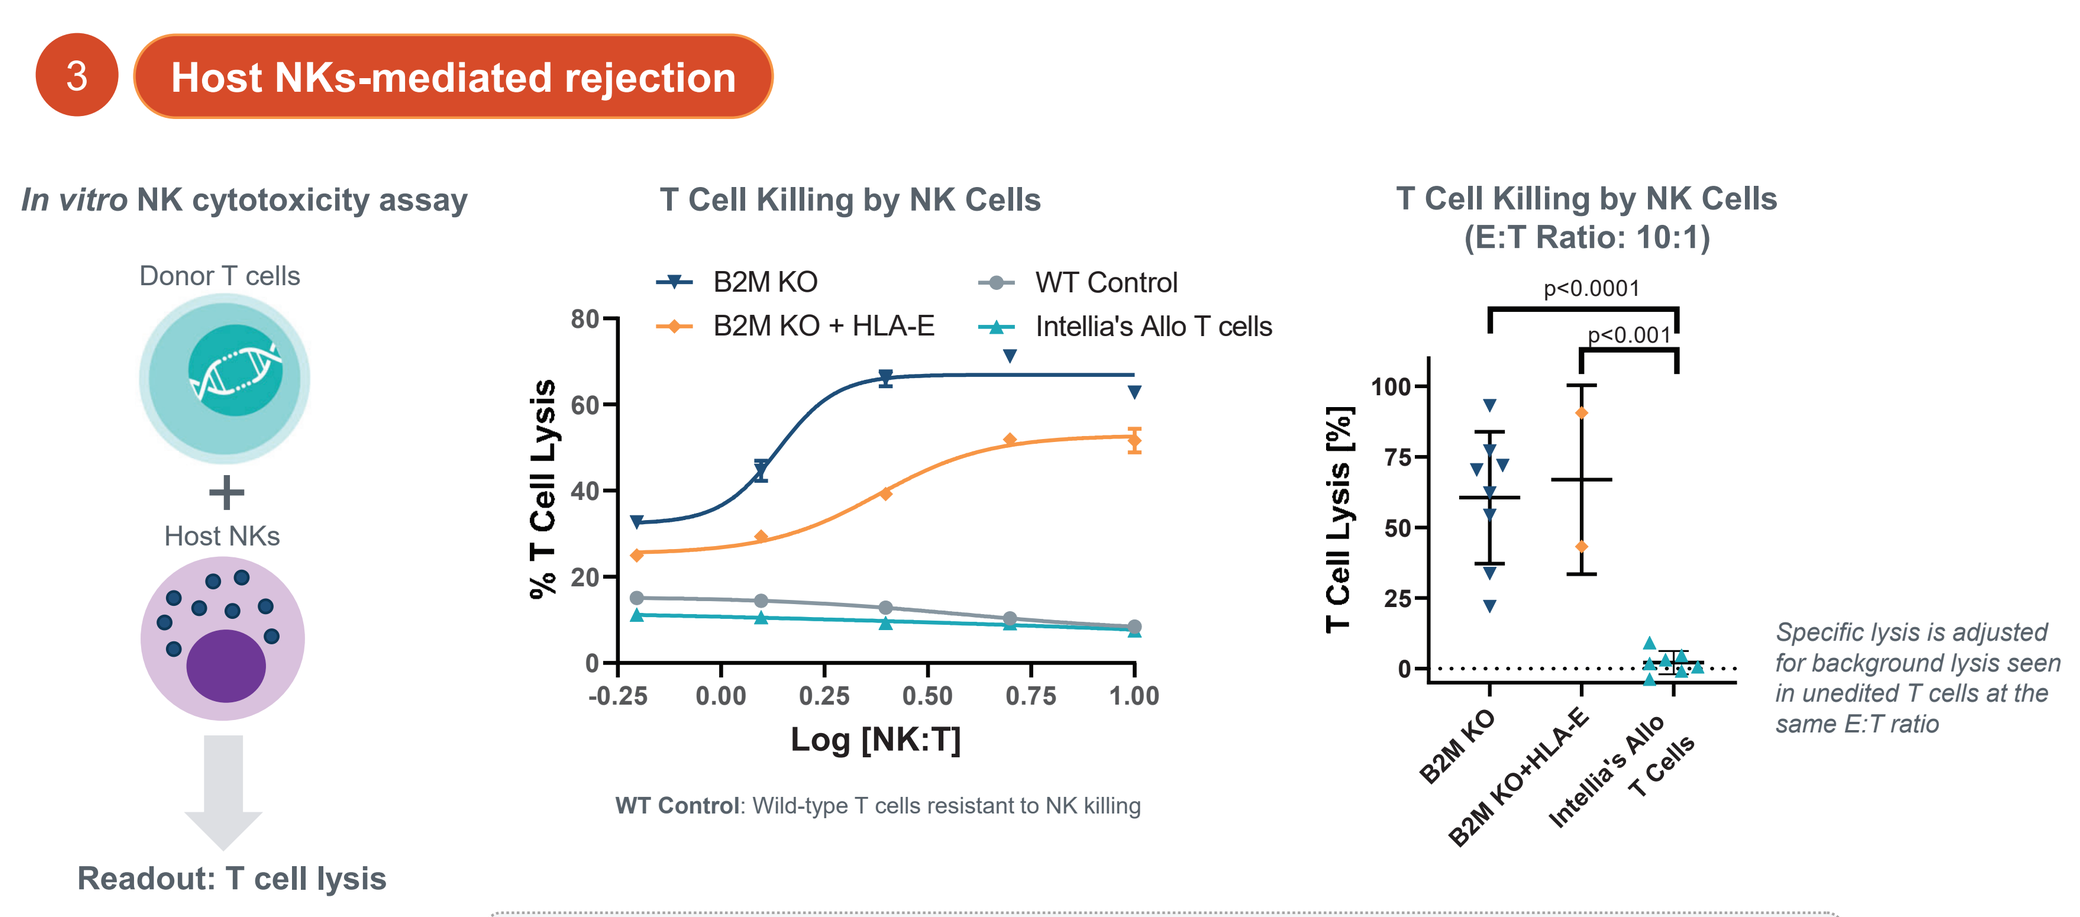
3 Host NKs-mediated rejection
In vitro NK cytotoxicity assay
Donor T cells
Host NKs
Readout: T cell lysis
T Cell Killing by NK Cells
B2M KO
B2M KO + HLA-E
WT Control
Intellia's Allo T cells
% T Cell Lysis
Log [NK:T]
WT Control: Wild-type T cells resistant to NK killing
T Cell Killing by NK Cells (E:T Ratio: 10:1)
p<0.0001
p<0.001
T Cell Lysis [%]
B2M KO
B2M KO+HLA-E
Intellia's Allo T Cells
Specific lysis is adjusted for background lysis seen in unedited T cells at the same E:T ratio

- Intellia's Allo T cells are protected from human NK cell elimination across multiple donors and HLA-B/C genotypes
- T cells lacking HLA class I are killed by NK cells; an effect only partially rescued by insertion of HLA-E

Allo T Cells Resisted NK Cell Killing for at Least 90 Days *in vivo*

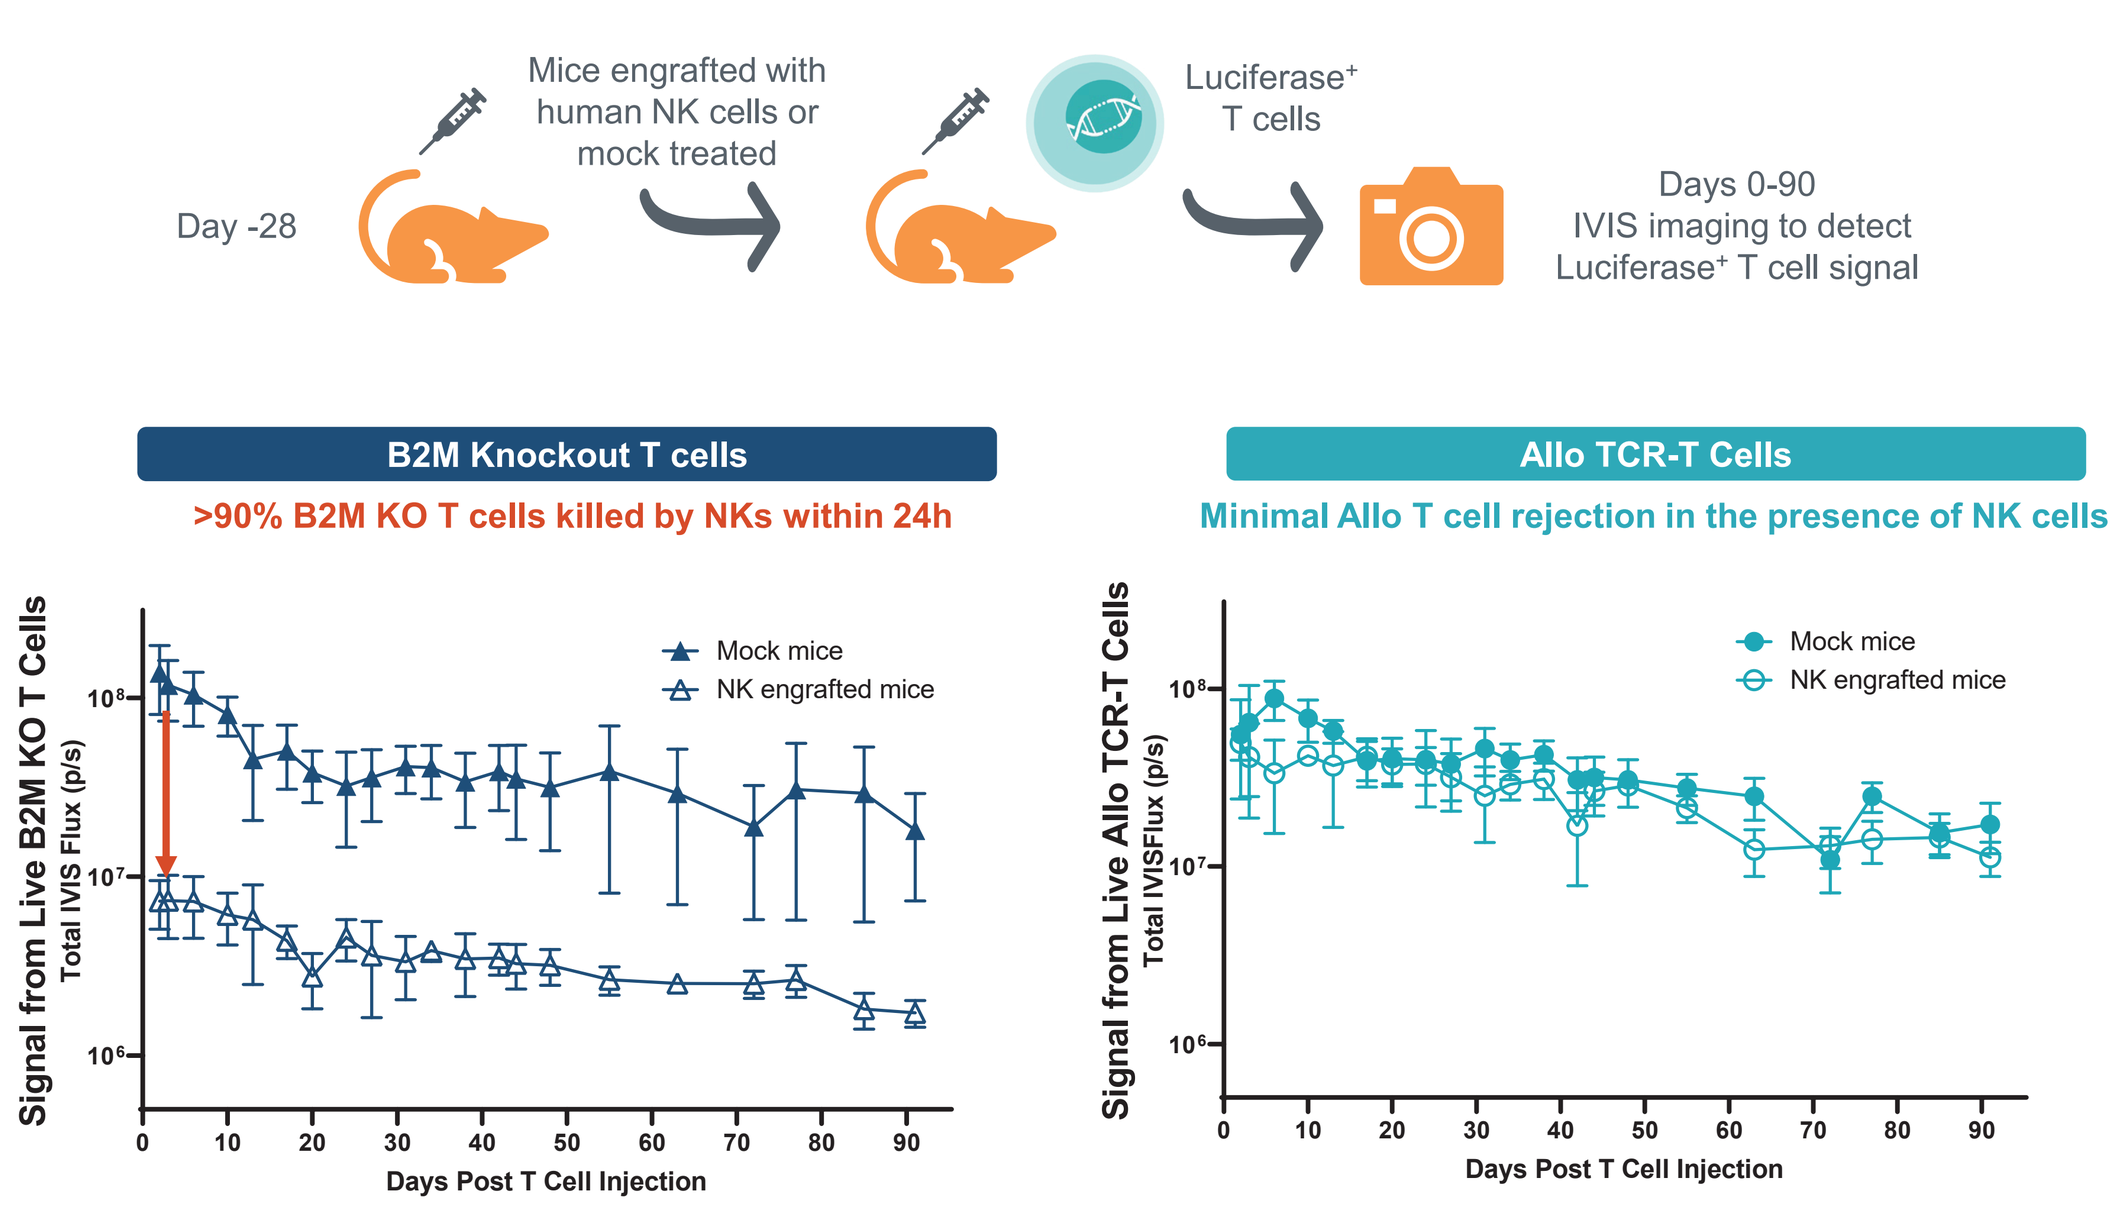
Day -28
Mice engrafted with human NK cells or mock treated
Luciferase+ T cells
Days 0-90
IVIS imaging to detect Luciferase+ T cell signal
B2M Knockout T cells
>90% B2M KO T cells killed by NKs within 24h
Mock mice
NK engrafted mice
Signal from Live B2M KO T Cells
Total IVIS Flux (p/s)
Days Post T Cell Injection
Allo TCR-T Cells
Minimal Allo T cell rejection in the presence of NK cells
Signal from Live Allo TCR-T Cells

HIGHLY EFFICIENT ALLOGENEIC CAR-T CELL ENGINEERING

Highly Efficient Allo CAR-T Cell Engineering at Bioreactor Scale (N=4 Donors)

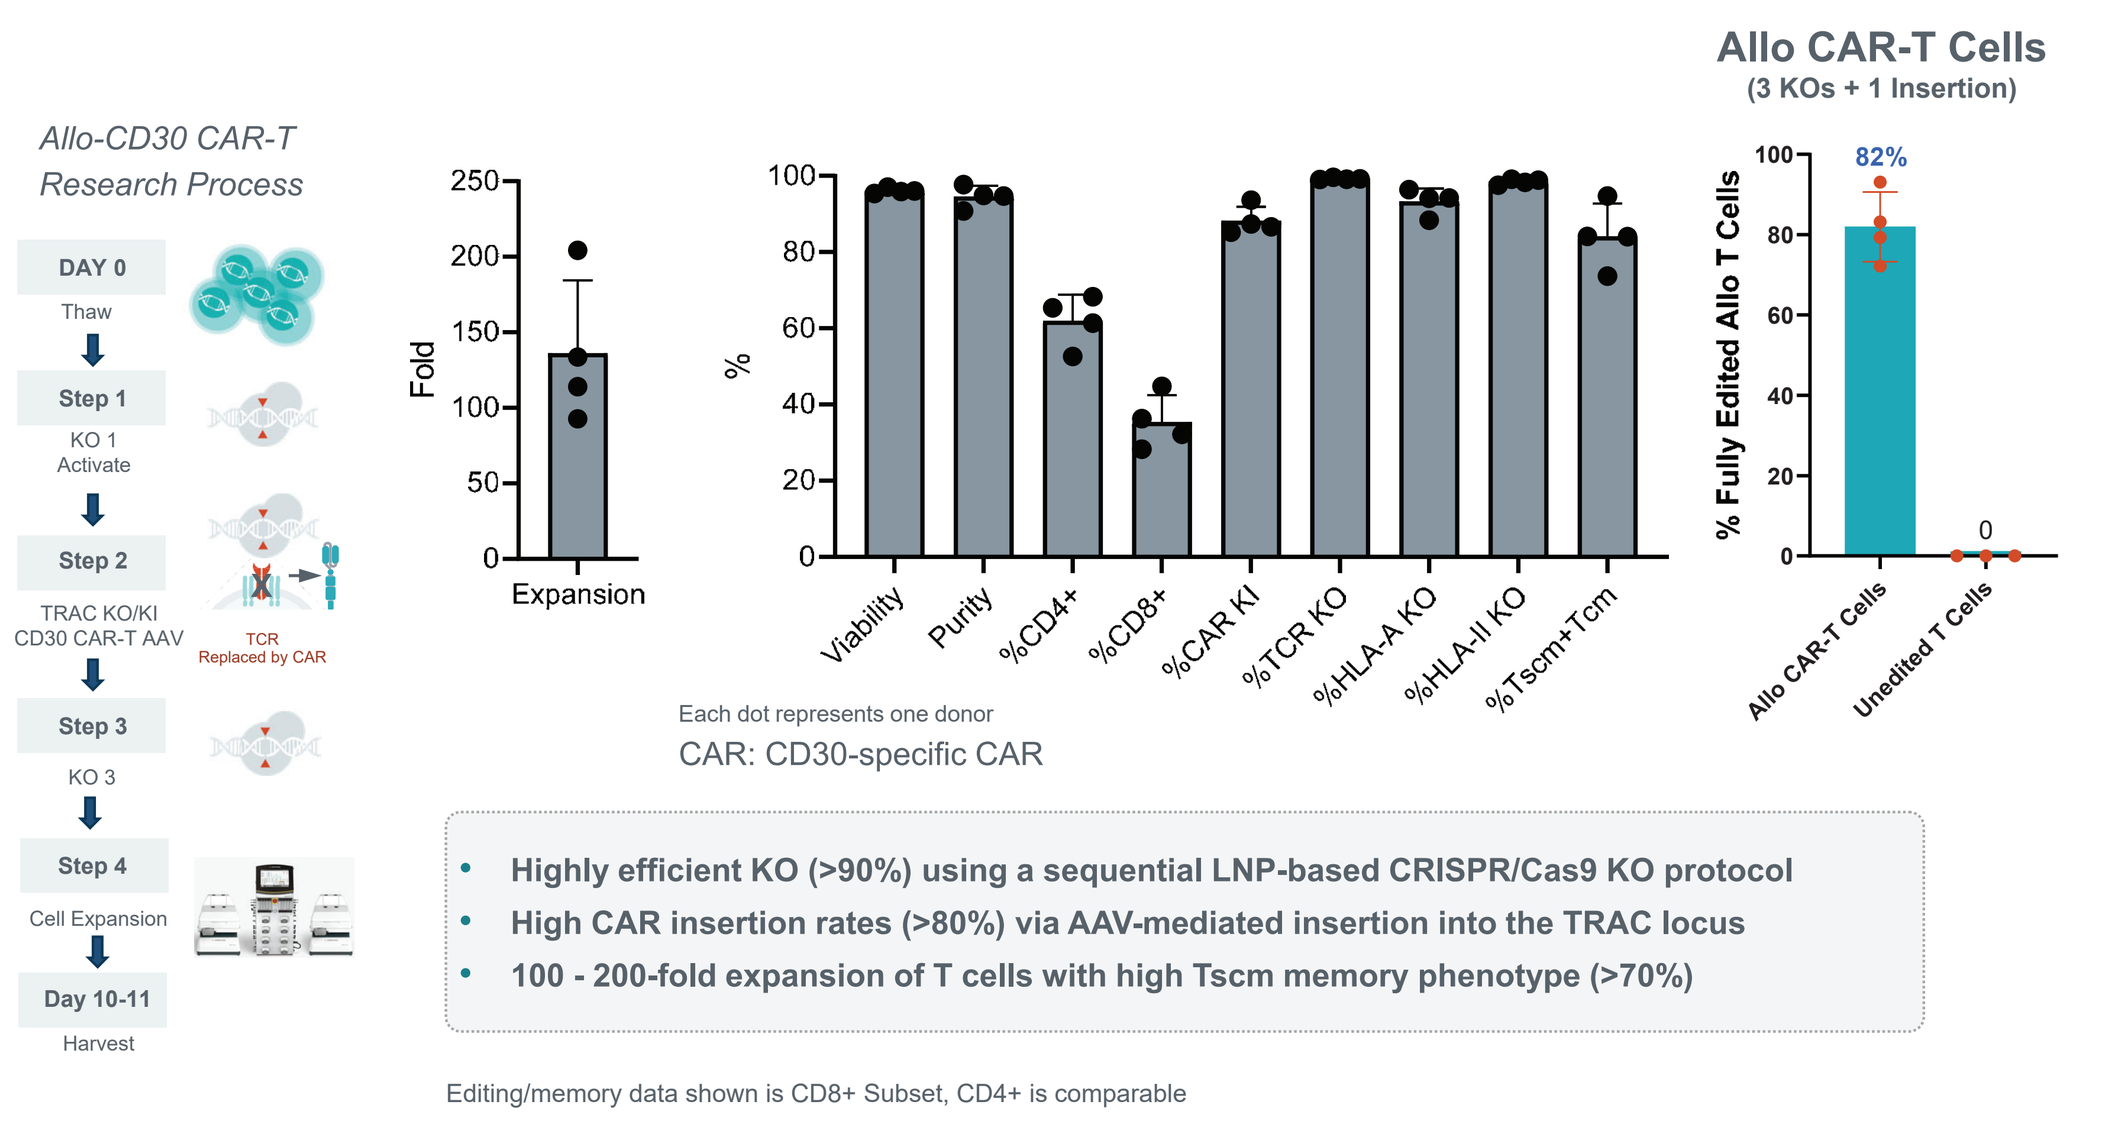
Allo-CD30 CAR-T Research Process
DAY 0
Thaw
Step 1
KO 1
Activate
Step 2
TRAC KO/KI
CD30 CAR-T AAV
TCR Replaced by CAR
Step 3
KO 3
Step 4
Cell Expansion
Day 10-11
Harvest
Fold
Expansion
Viability
Purity
%CD4+
%CD8+
%CAR KI
%TCR KO
%HLA-A KO
%HLA-II KO
%Tscm+Tcm
Each dot represents one donor
CAR: CD30-specific CAR
Allo CAR-T Cells
(3 KOs + 1 Insertion)
% Fully Edited Allo T Cells
82%
Allo CAR-T Cells
Unedited T Cells

- Highly efficient KO (>90%) using a sequential LNP-based CRISPR/Cas9 KO protocol
- High CAR insertion rates (>80%) via AAV-mediated insertion into the TRAC locus
- 100 - 200-fold expansion of T cells with high Tscm memory phenotype (>70%)

Editing/memory data shown is CD8+ Subset, CD4+ is comparable

Allo CAR-T Cells Have Comparable Anti-tumor Activity to Autologous T Cells

Example: CD30 CAR-T

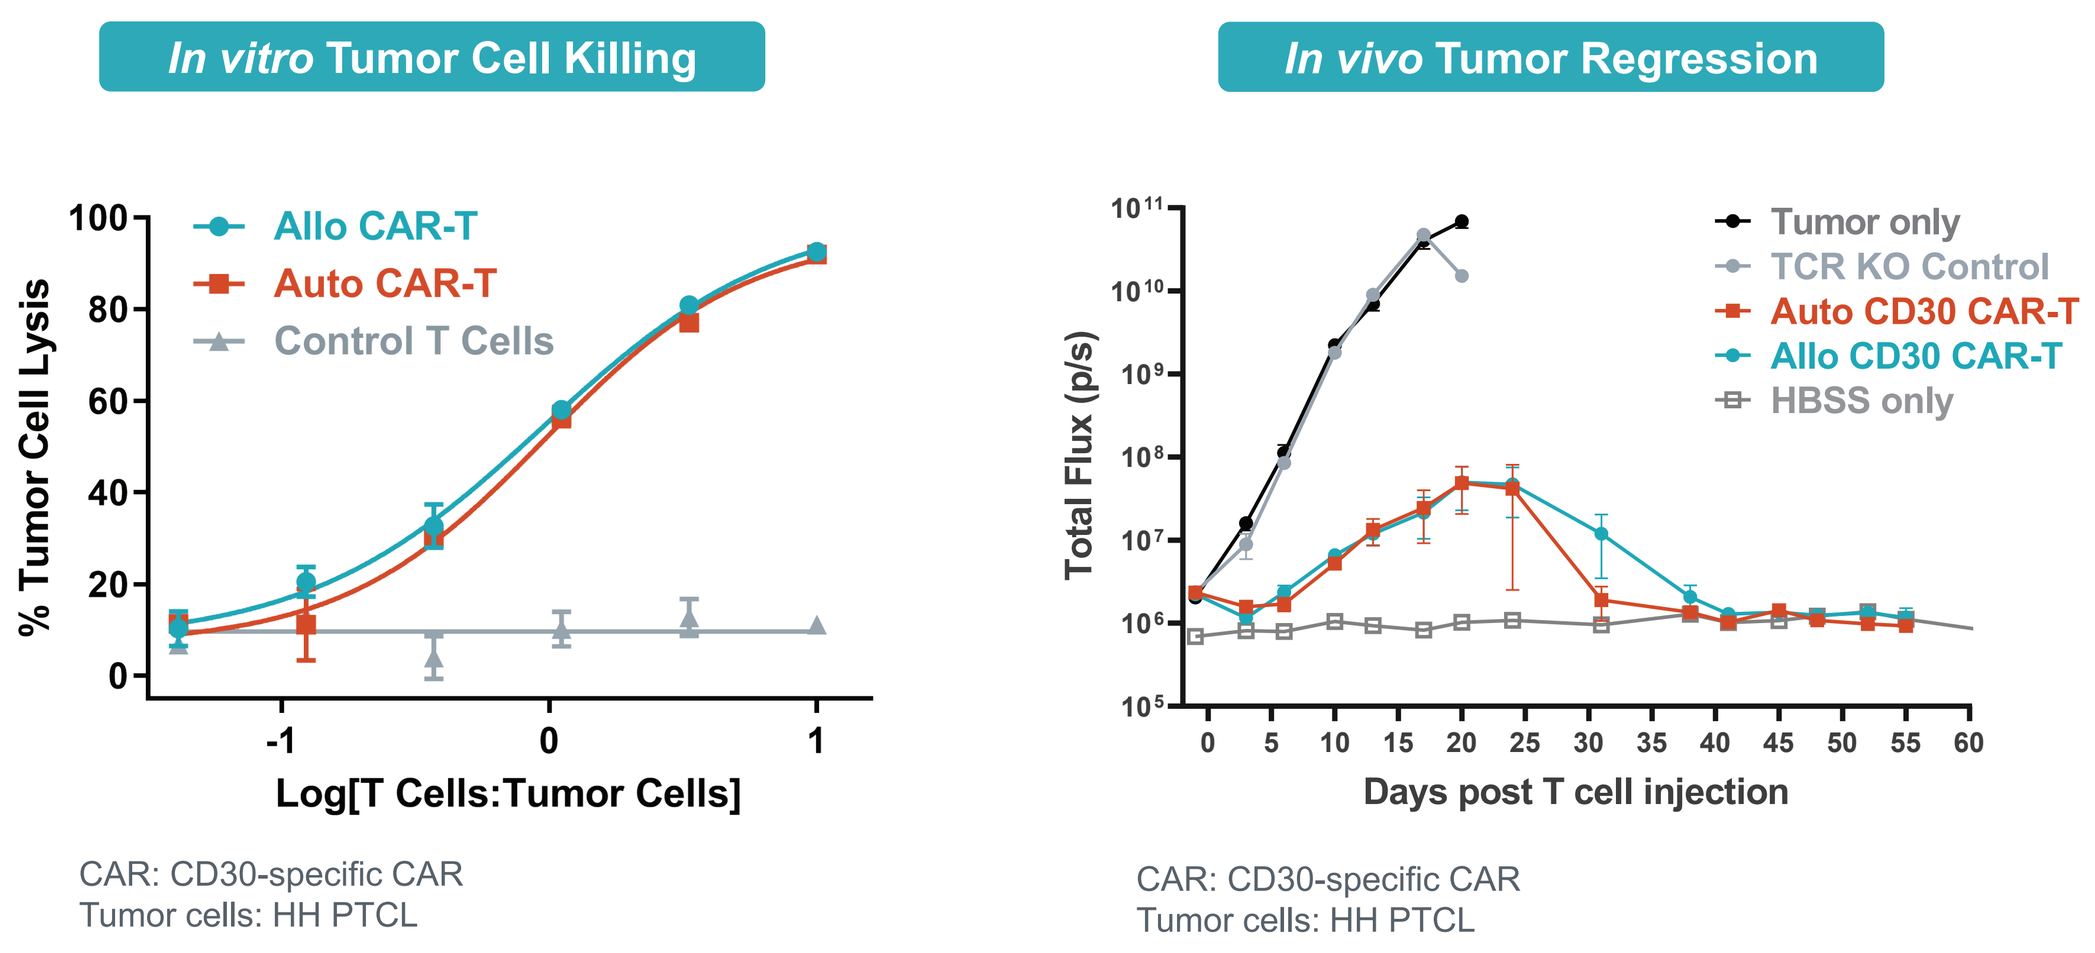
In vitro Tumor Cell Killing
Allo CAR-T
Auto CAR-T
Control T Cells
% Tumor Cell Lysis
Log[T Cells:Tumor Cells]
In vivo Tumor Regression
Tumor only
TCR KO Control
Auto CD30 CAR-T
Allo CD30 CAR-T
HBSS only
Total Flux (p/s)
Days post T cell injection

CAR: CD30-specific CAR
Tumor cells: HH PTCL

CAR: CD30-specific CAR
Tumor cells: HH PTCL

TCR-T CELL ENGINEERING

Allo-TCR-T Cells Are Efficiently Edited at all Loci Across Multiple Donors at Bioreactor Scale

- Highly efficient KO of HLA-A, CIITA KO (>95%)
- Near complete removal of the endogenous TCR
- High TCR insertion via AAV-mediated HR into *TRAC* locus
- >80% fully edited allo-TCR-T cells
- Predominant early memory phenotype
- Minimal T2T translocations

HR = homologous recombination
T2T = target-to-target
UED-T = un-edited T cells

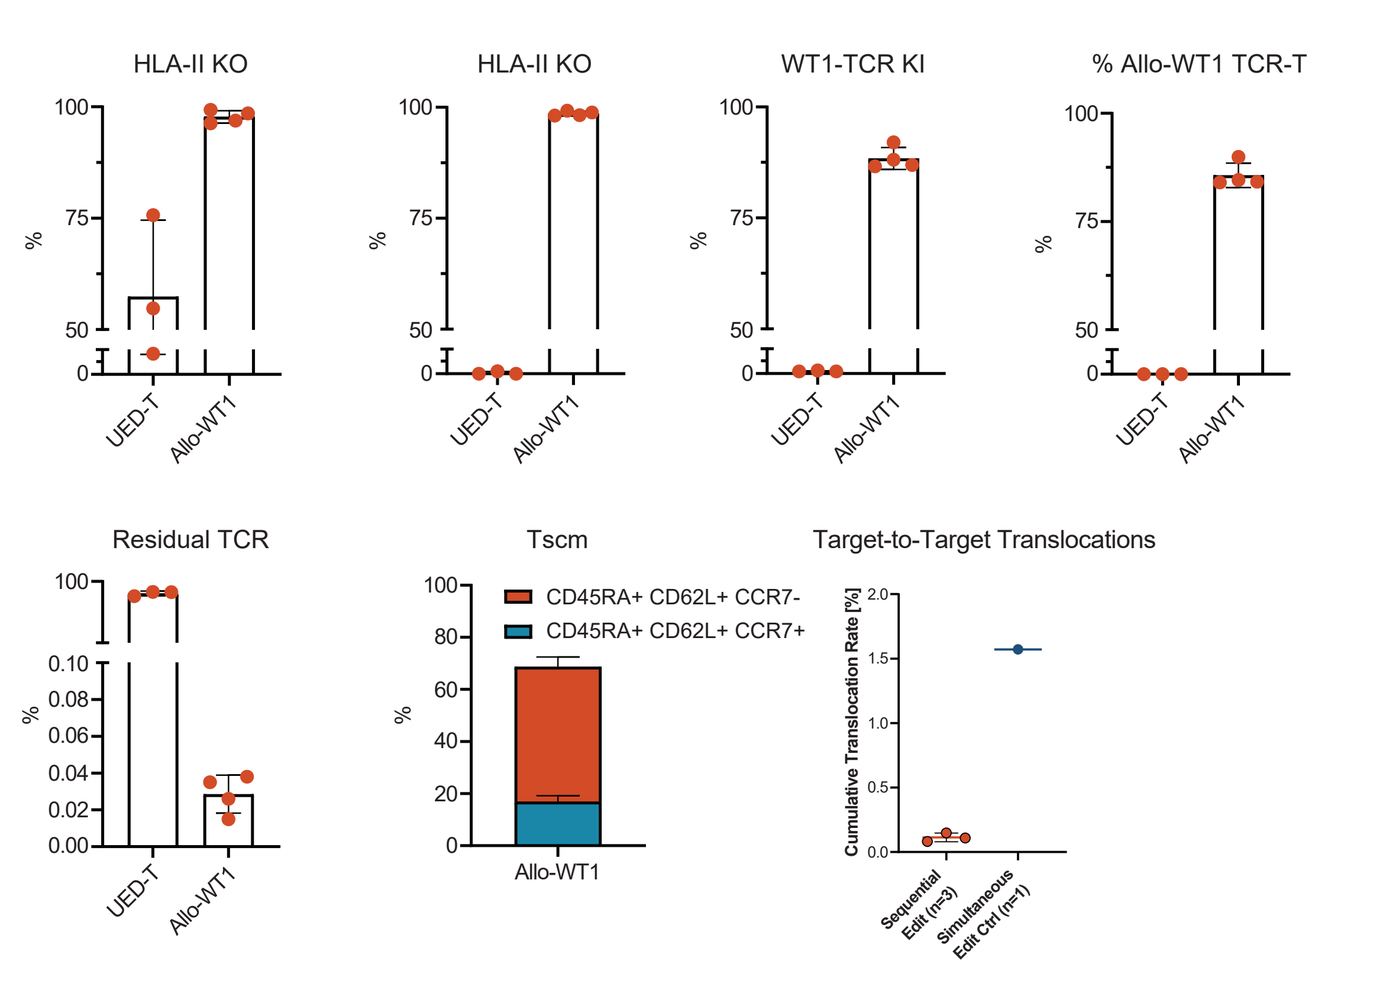
HLA-II KO
HLA-II KO
WT1-TCR KI
% Allo-WT1 TCR-T
UED-T
Allo-WT1
Residual TCR
Tscm
CD45RA+ CD62L+ CCR7-
CD45RA+ CD62L+ CCR7+
Target-to-Target Translocations

Allo TCR-T Cells Have Enhanced Anti-Tumor Activity over Autologous T Cells

Example: WT1 TCR

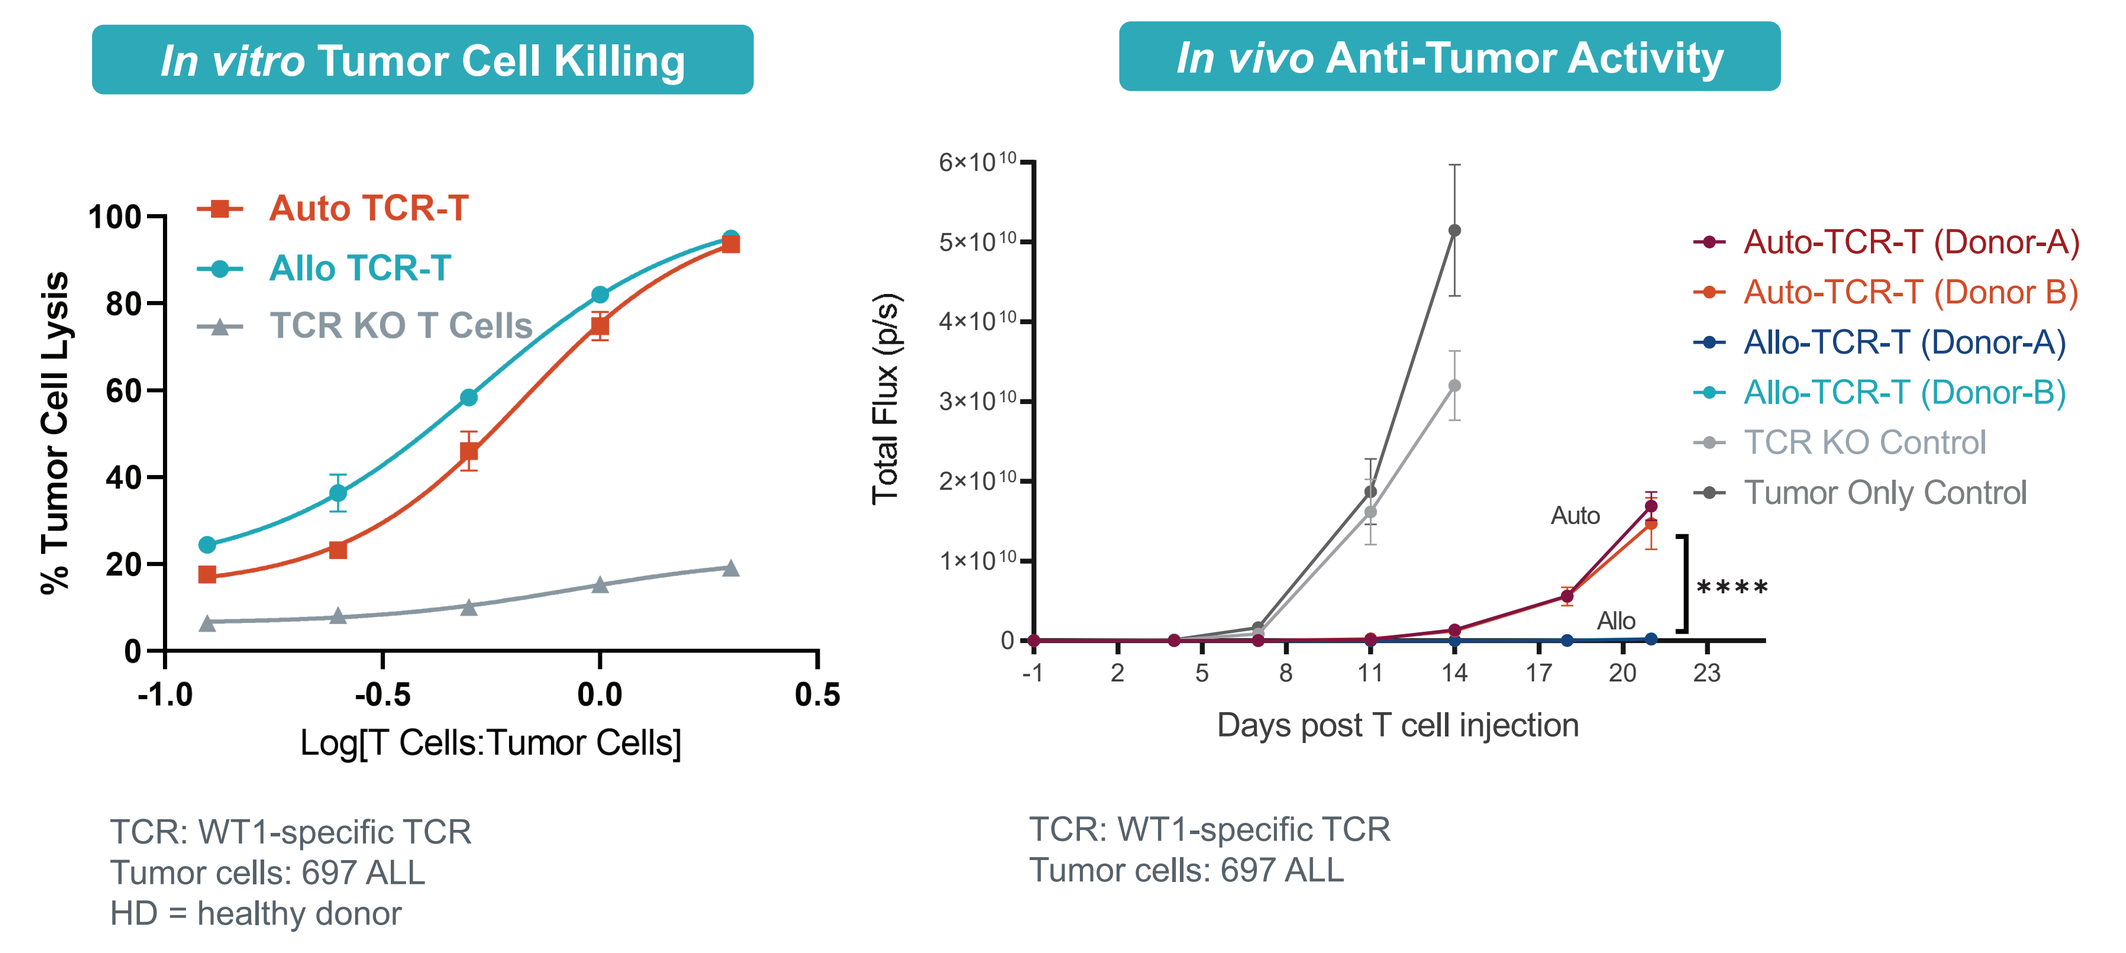
In vitro Tumor Cell Killing
Auto TCR-T
Allo TCR-T
TCR KO T Cells
% Tumor Cell Lysis
Log[T Cells:Tumor Cells]
In vivo Anti-Tumor Activity
Auto-TCR-T (Donor-A)
Auto-TCR-T (Donor B)
Allo-TCR-T (Donor-A)
Allo-TCR-T (Donor-B)
TCR KO Control
Tumor Only Control
Total Flux (p/s)
Auto
Allo

Days post T cell injection

TCR: WT1-specific TCR
Tumor cells: 697 ALL
HD = healthy donor

TCR: WT1-specific TCR
Tumor cells: 697 ALL

CONCLUSIONS

- **Intellia's differentiated allogeneic platform has shown key characteristics in *in vitro* assays and *in vivo* mouse models**
 - Eliminates GvHD concerns through highly efficient TCR knockout
 - Avoids recognition by host CD4 and CD8 T cells
 - Is protected from NK cell killing
- **HLA-B/C (2/2) matching strategy allows for small number (10-20) of donors to address majority of patients**
- **Approach is not expected to require intense immune suppression**
- **Allogeneic platform is readily deployable for TCR-T and CAR-T cell therapy**
 - Allo CAR-T cells showed no loss in anti-tumor activity to autologous counterparts in comparative *in vitro* and *in vivo* studies
 - A benefit was observed for allo-TCR-T cells
- **Nominated lead CD30 CAR-T as Intellia's first allogeneic development candidate (NTLA-6001)**

Intellia THERAPEUTICS